Abstract 5435 B759

Dopamine amacrine and interplexiform cells exhibit glutamatergic signatures

B.W. Jones, R.E. Marc, C.B. Watt • Univ Utah Moran Eye Center, Salt Lake City, UT • NIH EY02576

Purpose: Purpose: To investigate the small molecule signatures, physiologic responses and signaling mechanisms of vertebrate dopaminergic amacrine (AC) and interplexiform (IPC) cells.

Methods: Intrinsic small molecule signals were assayed along with *in vivo* and *in vitro* AGB permeation into retinal cells via computational molecular phenotyping (Marc and Jones 2002 J Neurosci 22: 413) in rabbit, mouse and goldfish retinas. Datasets were visualized as rgb and theme maps of classified cell types, thus reporting the identities and excitation states of all neuronal populations. We specifically visualized dopaminergic neurons via tyrosine hydroxylase (TH) signals, to unambiguously determine their small molecule phenotypes and mechanisms of responsivity.

Results: Mammalian TH+ ACs have been proposed to be GABAergic, yet we show that they all contain little or no GABA, low levels of glycine, and very high levels of glutamate. In this sense, they share the same molecular phenotype as goldfish IPCs. Rabbit TH+ ACs show moderately high levels of induced AGB permeation, indicating ionotropic glutamate receptor-mediated channel activation. This is consistent with evidence of low frequency OFF-center cone bipolar cell inputs to mammalian TH+ cells. In contrast, teleost TH+ IPCs show no endogenous AGB permeation, consistent with prior pharmacologic evidence that they are entirely under inhibitory control. TH+ ACs and IPCs resemble displaced ganglion cells, but express a molecular phenotype closer to bipolar cells (high glutamate + high taurine).

Conclusions: It is presumed that, but for the teleost dopaminergic IPC, all vertebrate amacrine cells are GABAergic or glycinergic (Marc et al., 1995; Kalloniatis et al., 1996), and that the mammalian TH+ AC is a dual GABA/dopamine cell (Contini and Raviola, 2003). We find that TH+ ACs and IPCs possess a definitive glutamatergic signature resembling the vertical channels of the retina. All vertebrate dopaminergic retinal neurons are strongly activated by release-from-inhibition and may function in part by increasing tonic excitatory drive via synaptic glutamate release.

Commercial Relationship: BW Jones, None; CB Watt, None; RE Marc, Signature Immunologics F, E.

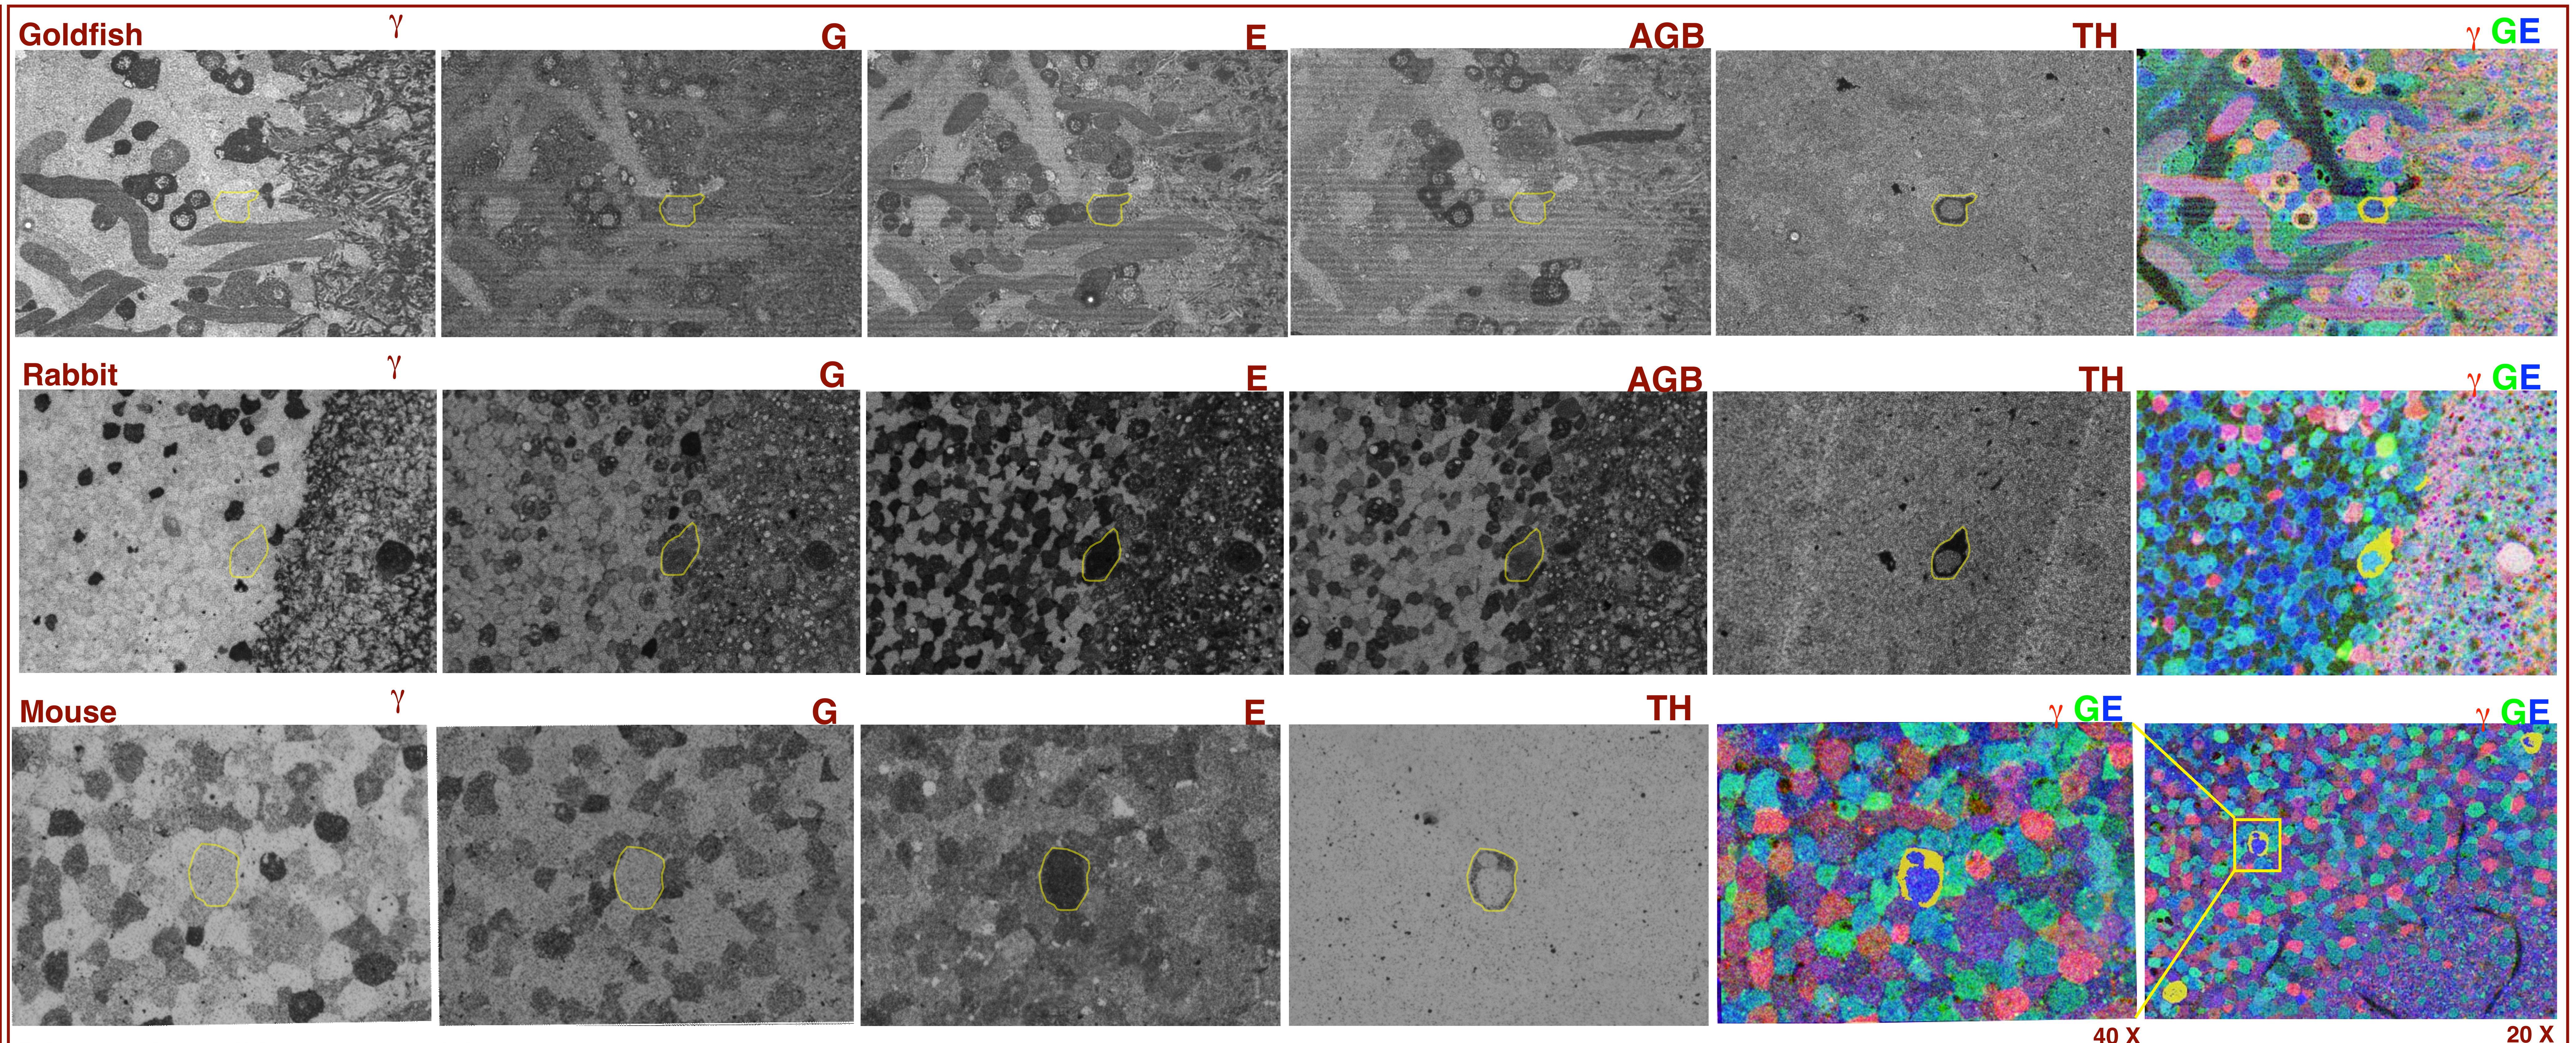
Goldfish
γ
G
E
AGB
TH
γ GE
Rabbit
γ
G
E
AGB
TH
γ GE
Mouse
γ
G
E
TH
γ GE
γ GE
40 X
20 X

DA amacrine cell immunoreactivity

Grayscale images of GABA, glycine, glutamate, and tyrosine hydroxylase immunoreactivity in goldfish, rabbit and mouse horizontal sections of retina. Goldfish and rabbit are also shown with glutamate mediated activation as revealed by AGB permeation. All color images to the right are RGB reconstructions with GABA, glycine and glutamate assigned to red, green and blue color channels respectively. AGB signaling in the goldfish and rabbit reveals differential uptake of AGB based upon likely differing cohorts of glutamatergic channels or channel mediated receptors with TH amacrine cells in rabbit being much more responsive to glutamatergic signaling than the goldfish TH amacrine cell. TH+ amacrine cells are also shown in lower right to be at the expected densities of ~10/mm^2 in the peripheral retina (Dacey 1990).

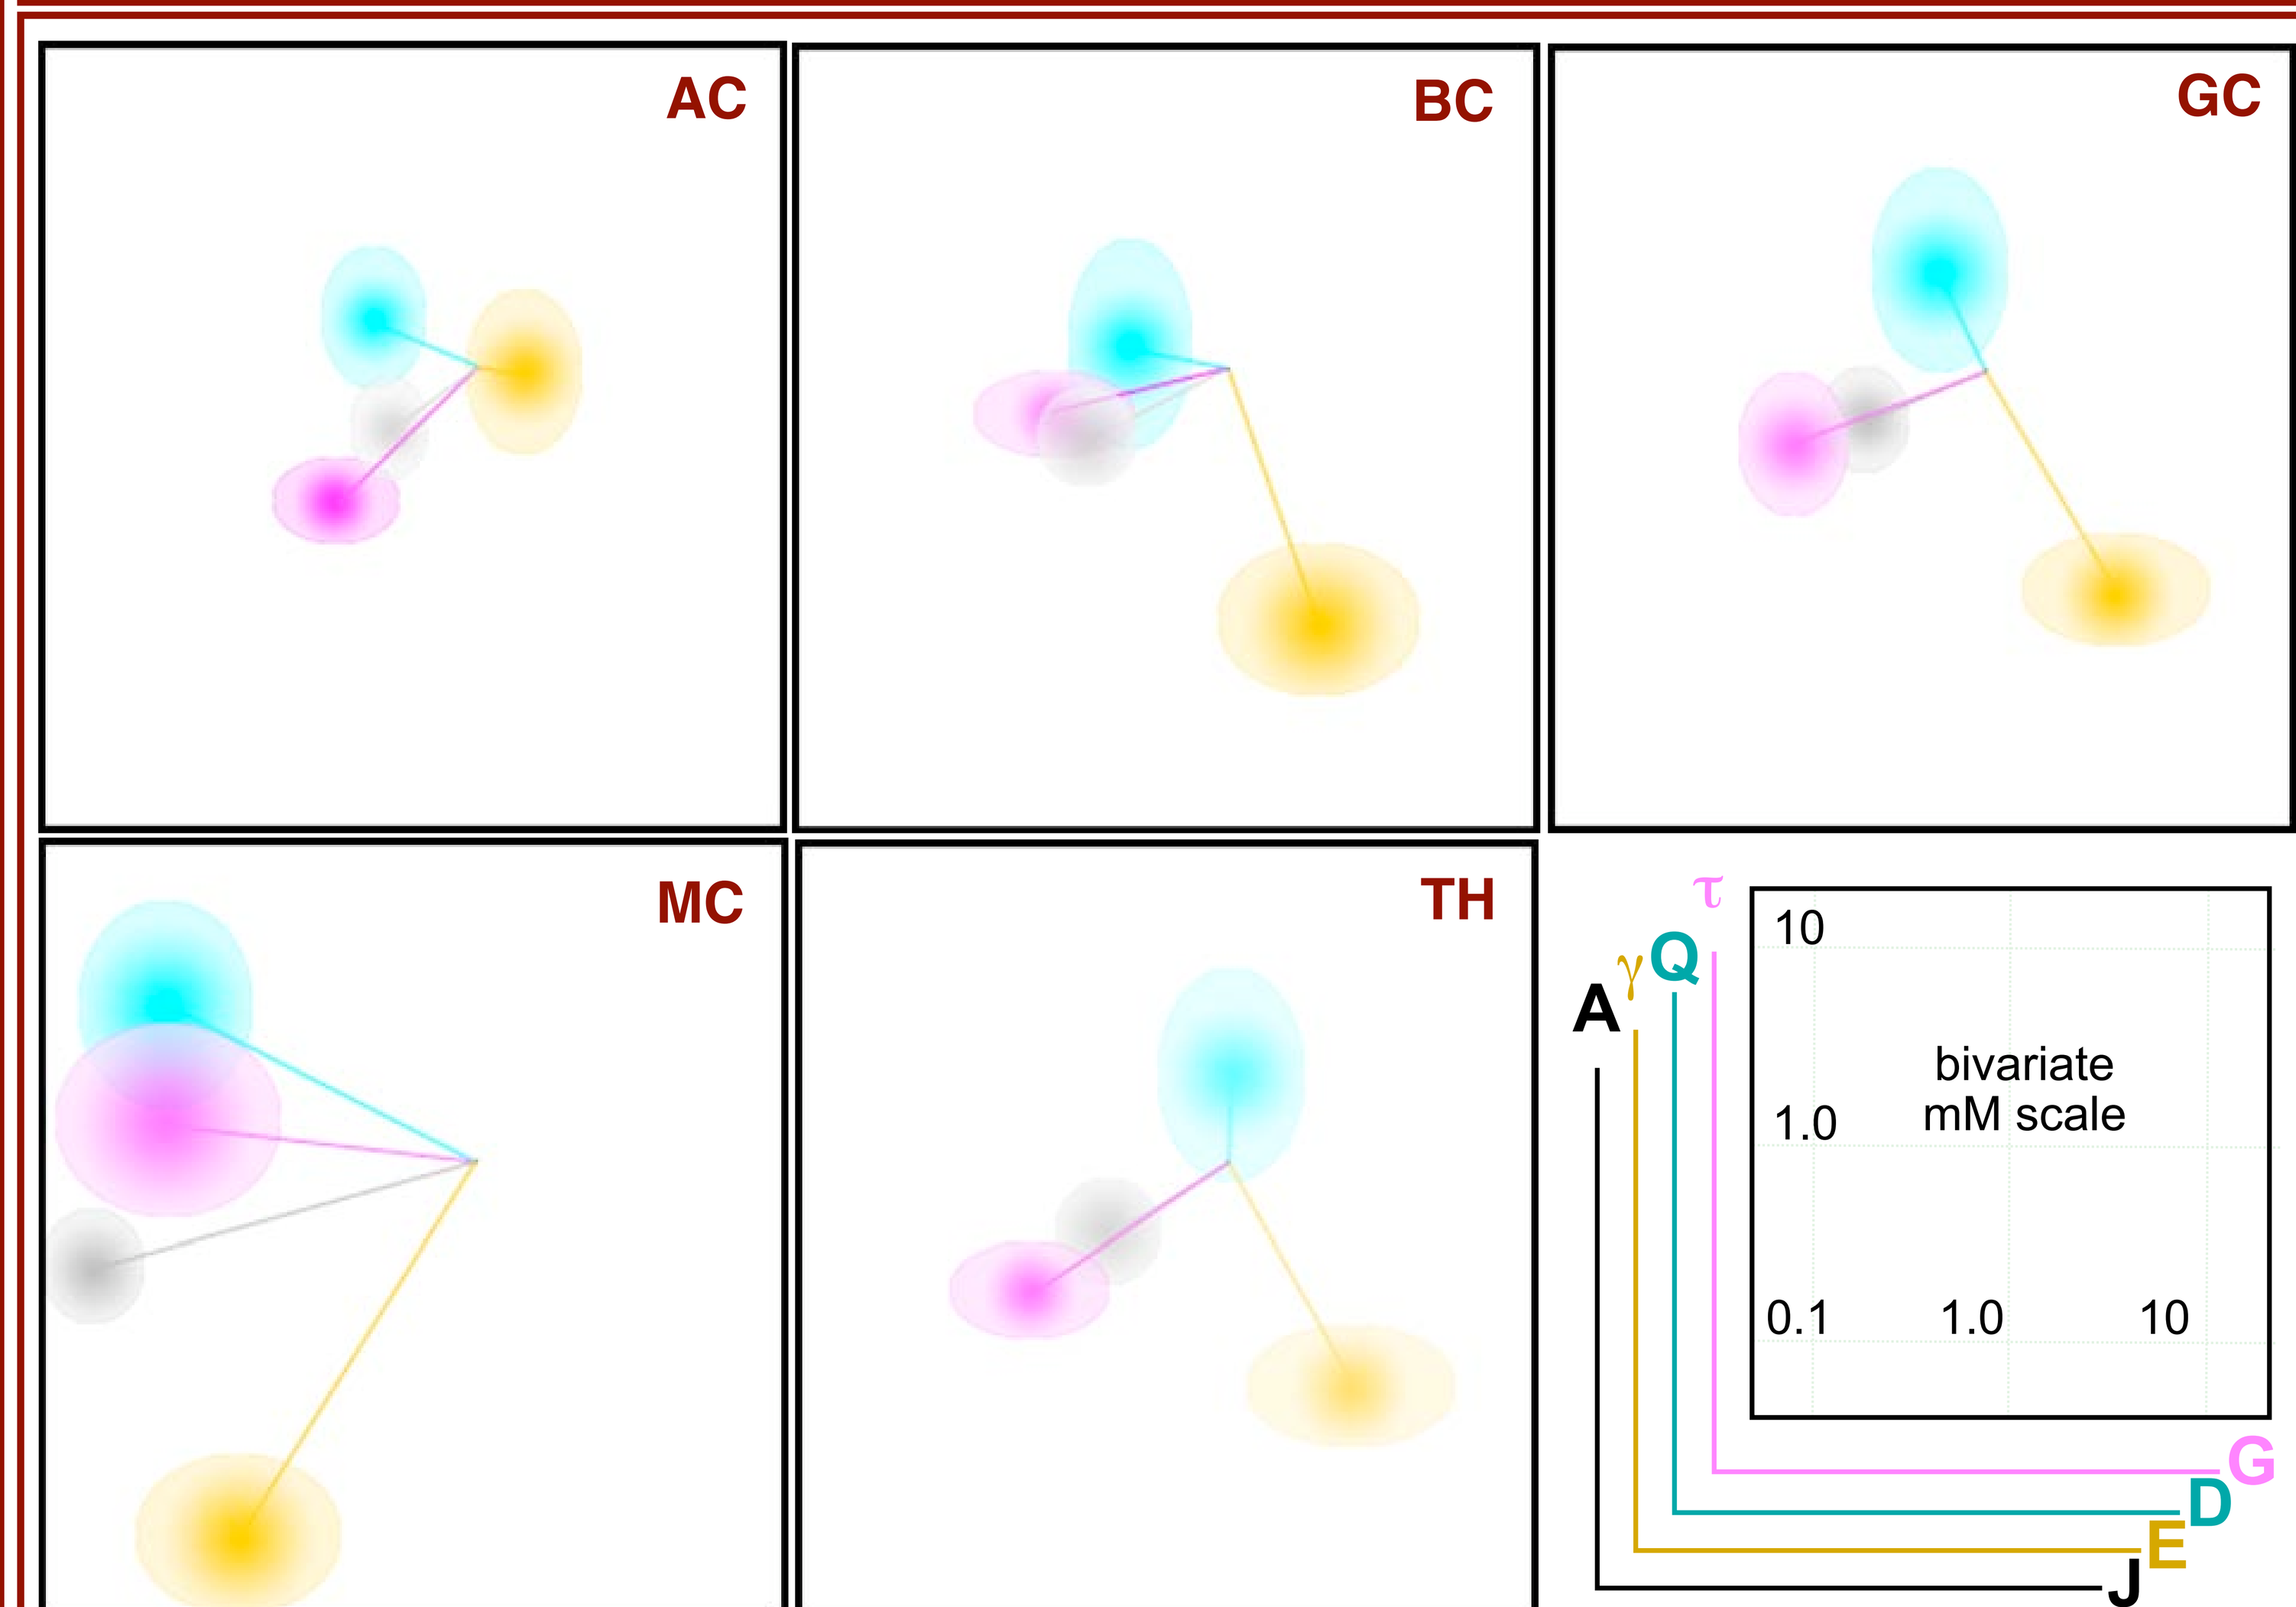
AC
BC
GC
MC
TH
A
γ
Q
τ
10
1.0
0.1
1.0
10
bivariate
mM scale
G
D
E
J

Mouse small molecular phenotypes: Each class displays a unique molecular phenotype visualized as an N-dimensional phenotypic map

Each cell super class (AC=Amacrine cells, BC=Bipolar cells, GC=Ganglion cells, MC=Müller cells, TH=Tyrosine hydroxylase immunoreactive amacrine cells) in these graphs is represented by 4 superimposed bivariate density plots. Each data cloud is delimited by 2 SD borders. The x,y axis pairs are glutathione, alanine (JA); glutamate,GABA (Eγ); aspartate, glutamine (DQ); glycine, taurine (Gτ); spanning 0.1 - 10 mM. Vertical channel pathway cells (ganglion cells, bipolar cells) are defined by a specific small molecular phenotype described by a high glutamate signal and a glutamine concentration similar to or much higher than the glutamate content. Amacrine cell classes have been plotted as one super class encompassing all amacrine cell classes but overall can be described by very high GABA concentrations and relatively high glutamine contents. Glycine also is present in relatively large concentrations overall, but this level betrays some amacrine cell classes possessing very high levels of glycine. Müller cells are characterized by low concentrations of glutamate, no GABA, no glutathione, but high amounts of glutamine and taurine. From these plots, it can be seen that the TH positive amacrine cells possess a small molecular profile that more closely matches that of the vertical excitatory channel neurons that it does of any other cell class in the retina. What is not shown in these plot profiles is the degree of similarity TH+ amacrine cells hold with respect to bipolar cell neurons. Bipolar cell neurons express high taurine levels similar to that found in the TH+ amacrine cells which is in direct contrast to the absent in rabbit and mouse to very low levels found in mouse ganglion cells.