Abstract 2487 B434

Structure and Function of Microneuromas in Retinal Remodeling

BW Jones[1], RE Marc[1], CB Watt[1], K. Kinard[1], D. DeMill[1], J-H. Yang[1], T. Tasdizen[2], P. Koshevoy[2], E. Jurrus[2], R. Whitaker[2] • Moran Eye Center[1] • School of Computing/Scientific Computing Institute[2], Univ of Utah, Salt Lake City, UT

NIH EY02576 (RM), EY015128 (RM), EY014800 Vision Core (RM), EB005832 (TT), & Research to Prevent Blindness (BWJ & RM)

Purpose: Retinal remodeling triggered by retinal degenerations can lead to formation of new synaptic neuropil: microneuromas. The goals of our work are to visualize the fine structure, circuitry and functional attributes of microneuromas.

Methods: Models used include the human rhodopsin-GFP knock-in mouse (J Wilson, Baylor Col Med), an RGS9 truncation transgenic mouse (Jason C-K Chen, Virginia Commonwealth Univ) and the rd1 mouse. Postnatal day 100-450 mice were euthanized, eyes enucleated and fixed for visualization by computational molecular phenotyping (CMP; Jones et al., J Comp Neurol 2006;464: 1-16) or incubated for *in vitro* excitation mapping (Marc, *J Comp Neurol* 1999; 407:47-64) using 1-amino-4-guanidobutane (AGB) permeation activated by kainate or NMDA, followed by CMP. Some mice were used for *in vivo* AGB mapping of endogenous activity with 5 mM AGB in the eyecup for 45 min. Reconstructions of microneuromas were achieved by a combination of CMP and large-scale image tiling, registration and process tracking of serial high-resolution electron microscope imagery of microneuromas.

Results: Reconstructions of microneuromas reveal bipolar, amacrine and ganglion cells. Though dominated by conventional GABAergic synapses, bipolar cells form abundant synaptic ribbon contacts with all classes of profiles in microneuromas. Microneuromas are partitioned into distinct structural zones: (1) Muller process ensheathment; (2) orderly fascicles of *en passant* processes that make few or no contacts; (3) tangles of processes forming numerous synaptic connections. Two reconstructed microneuromas demonstrate either direct contact with remnant retinal pigmented epithelial (RPE) cells or RPE root-like processes extending into the core of the microneuroma. Excitation mapping with kainate or NMDA activation *in vitro* demonstrates that microneuromas express abundant functional ionotropic glutamate receptors. *In vivo* excitation mapping shows that microneuromas have intrinsic excitation events, even in the absence of photoreceptor drive.

Conclusions: Microneuromas are complex structures with intrinsic neural derived processes from cells that express functional ionotropic glutamate receptors. Microneuroma formation is potentially triggered by contact with the RPE. As some processes in microneuromas derive from retinal ganglion cells, ectopic signaling complicates attempts to restore visual signaling with transplants or prosthetic devices.

Commercial Relationship: BW Jones, None; RE Marc, Signature Immunologics; CB Watt, None; K Kinard, None; D DeMill, None; J-H Yang, None; T Tasdizen, None; P Koshevoy, None; E Jurrus, None; R Whitaker, None.

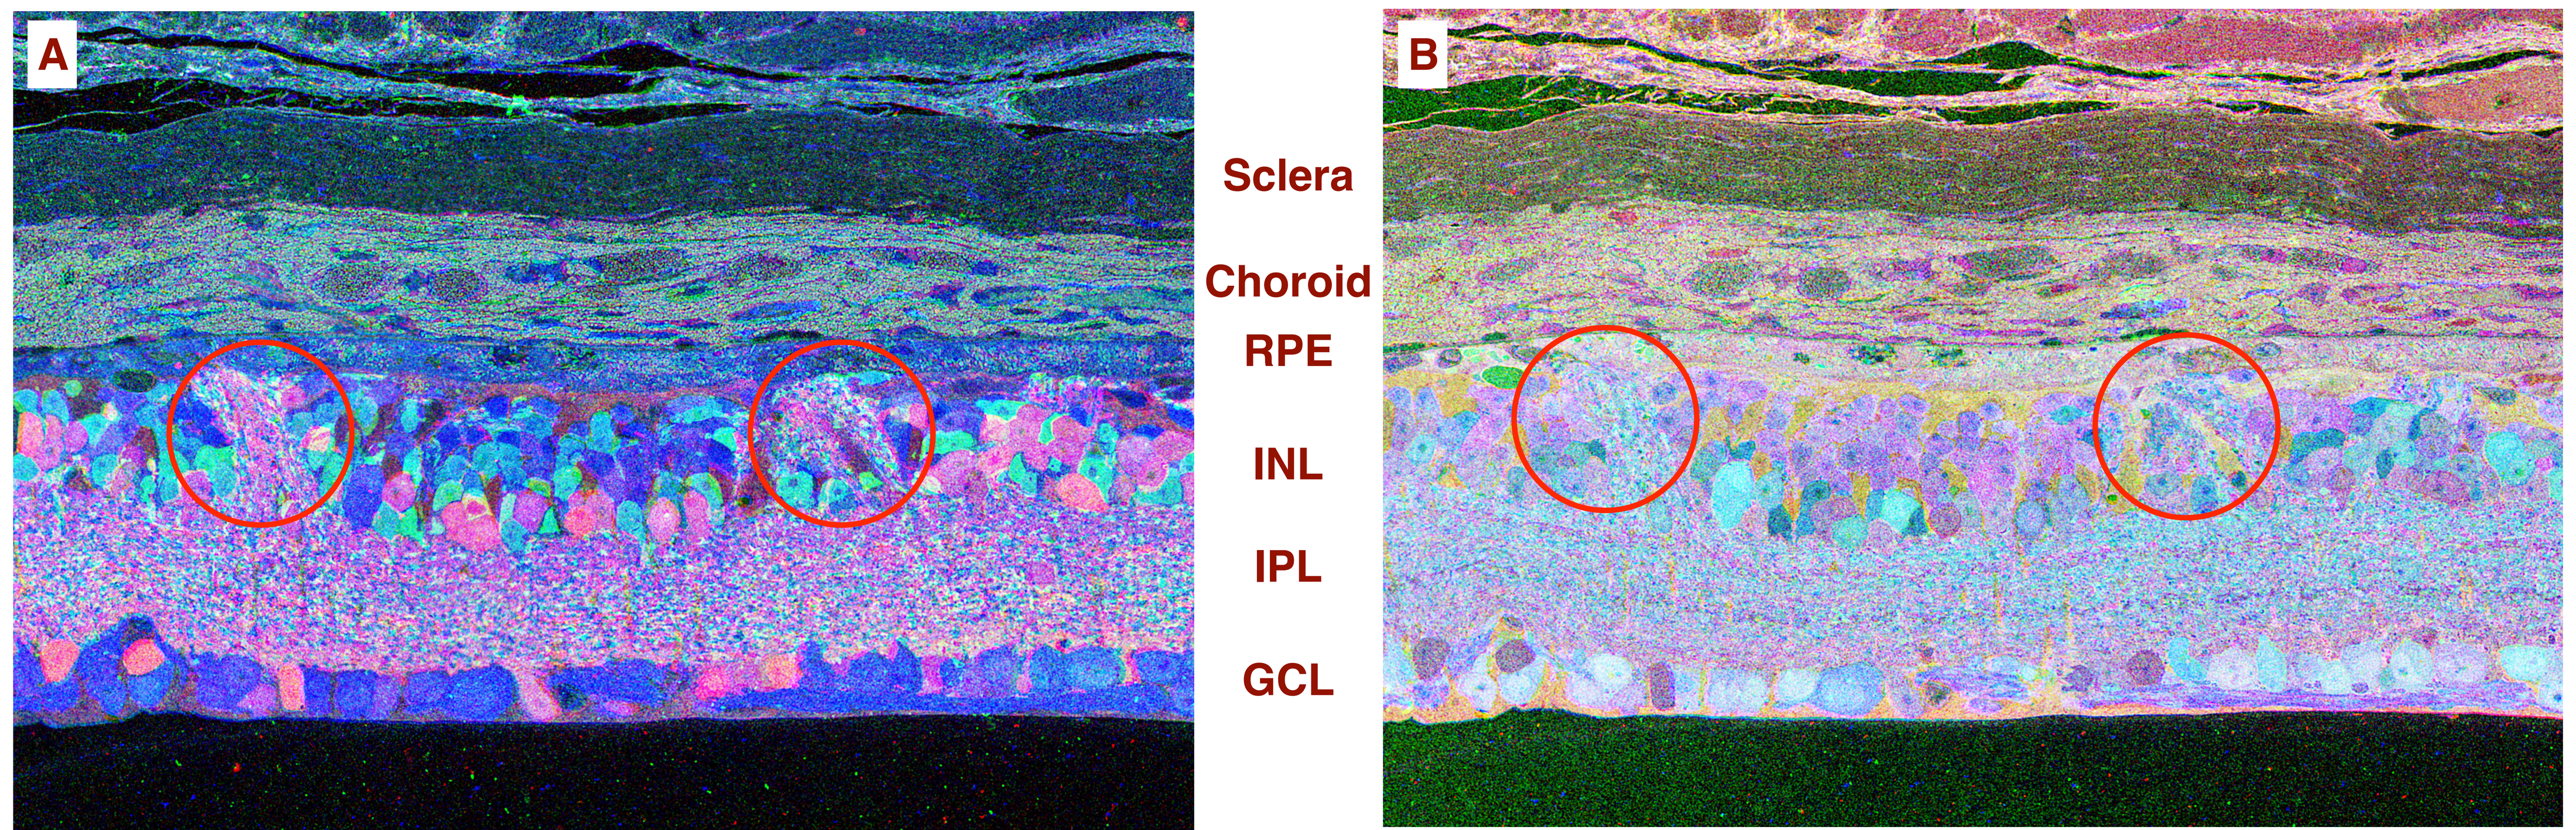
A
B
Sclera
Choroid
RPE
INL
IPL
GCL
γGE → rgb
τQE → rgb

Figure 1 Microneuroma columns in the human rhodopsin-GFP knock-in mouse shown in Fig. 1A. GABA (γ), glycine (G), glutamate (E) to RGB mapping and in Fig. 1B, taurine (τ), glutamine (Q), glutamate (E) to RGB mapping on the right. Microneuromas are tangles of GABAergic amacrine cell, glycinergic amacrine cell, glutamatergic bipolar cell and ganglion cell processes, ranging from 20 to over 100 μm in diameter and exceeding 30,000/retina in some models (GHL, TG9N, RCS and hrhoG). It is likely that all cell populations participate in microneuroma formation including horizontal cells. This novel neuropil formation develops in response to de-afferentation of the neural retina as a result of photoreceptor loss and is most aggressive in models that have coherent photoreceptor loss with rod and cone dystrophy. Additionally, it should be noted that microneuromas are common with potentially thousands of microneuromas per square millimeter in the remnant inner nuclear layer in mid stage retinal degeneration before massive cell death and even cell emigration out of the retina into the remnant choroid in late stage retinal degenerative disease.

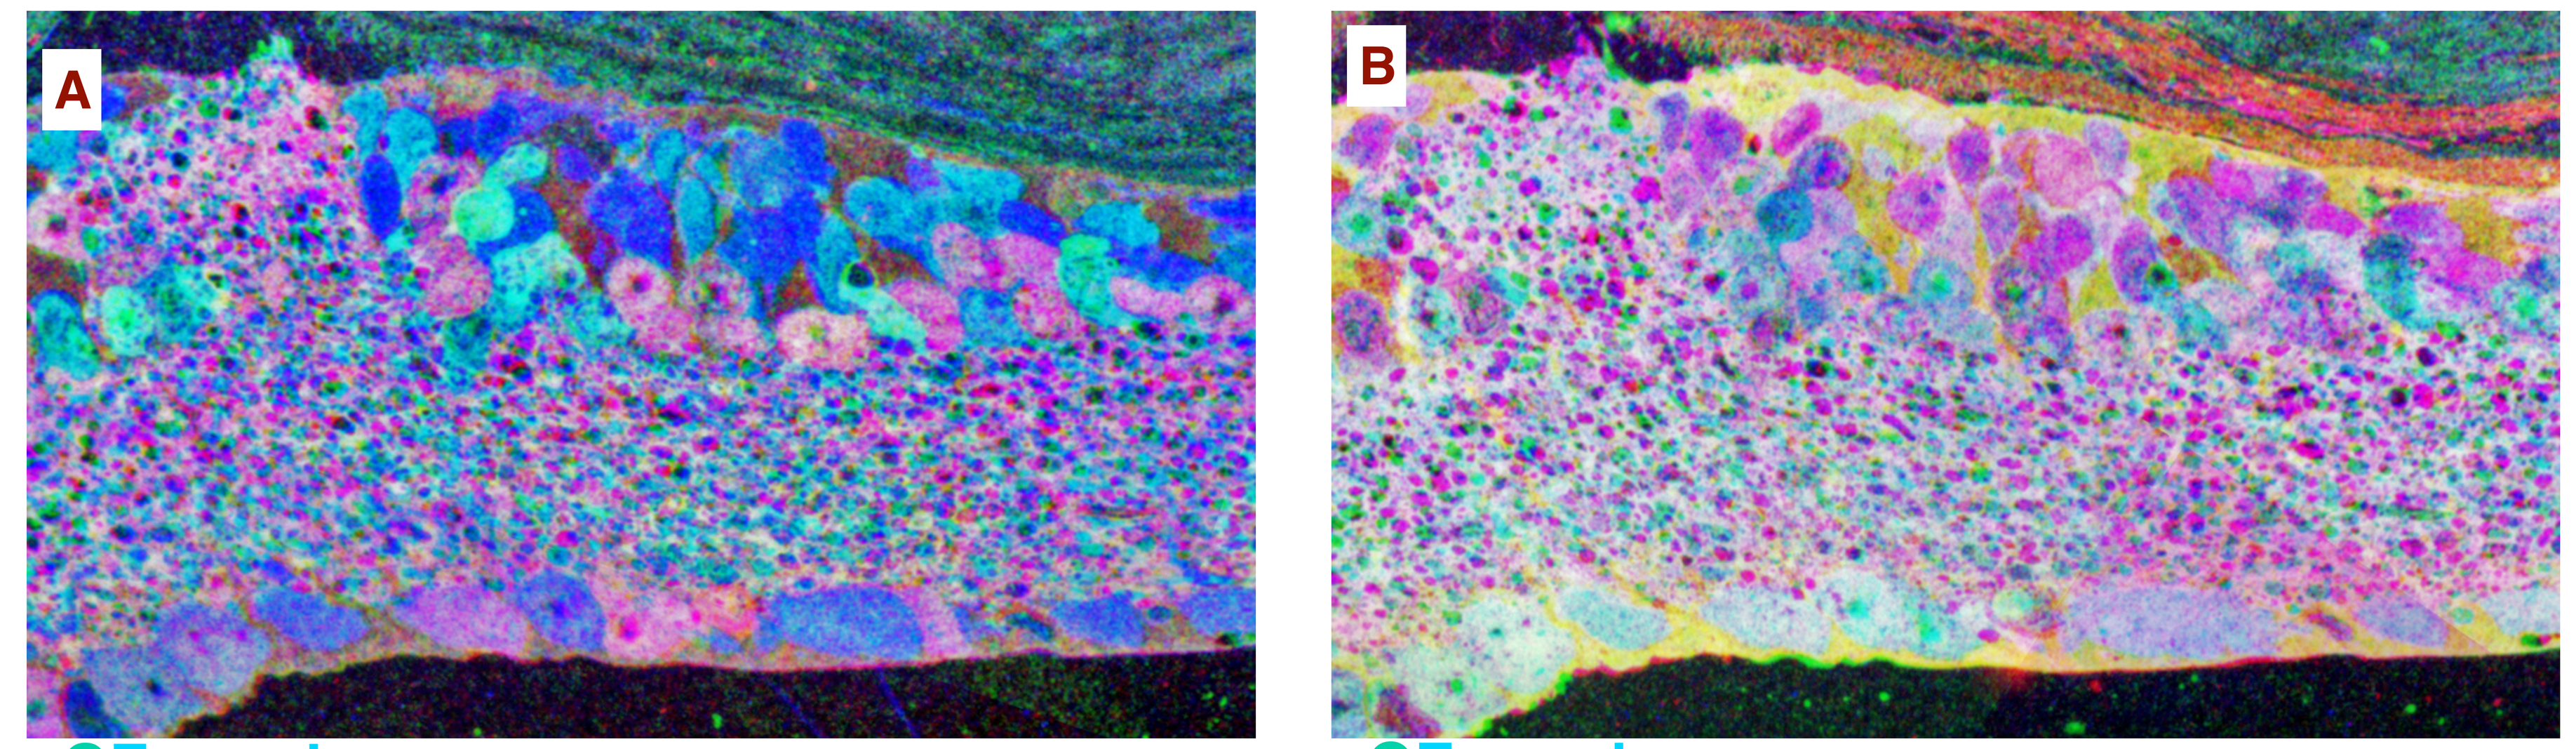
A
B
γGE → rgb
τQE → rgb

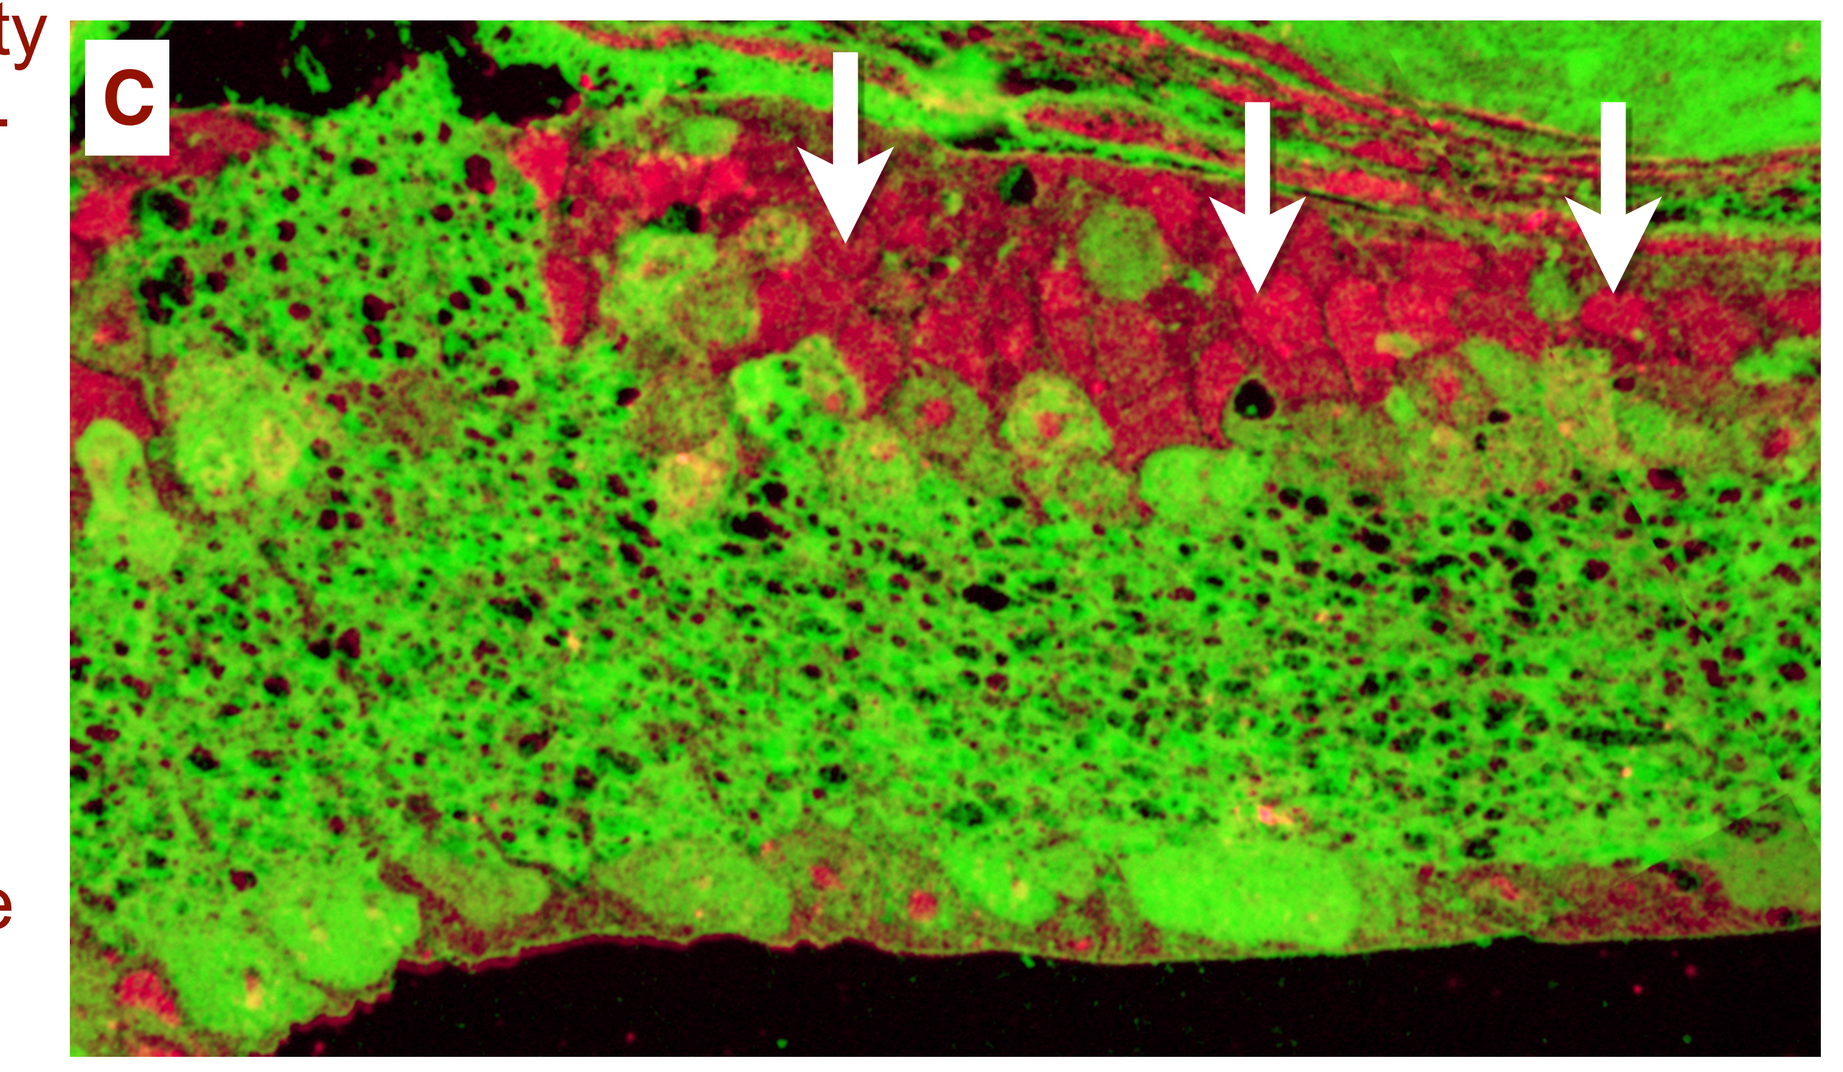
C
TB_AGB_x → rgb

Figure 2 Examination of glutamatergic mediated activity in microneuromas of the human rhodopsin-GFP knock-in mouse shown with 1A: GABA (γ), glycine (G), glutamate (E) to RGB mapping. Fig. 1B: taurine (τ), glutamine (Q), glutamate (E) to RGB mapping and 1C showing toluidine blue, AGB, x to RGB mapping. *In vitro* excitation mapping in retinas activated by KA reveals functional ionotropic glutamate receptor expression by amacrine and ganglion, but not bipolar cells (arrows). Importantly, processes in the microneuroma are extremely active, consistent with the large number of bipolar ribbons in them. In vivo excitation mapping (not shown) demonstrates intrinsic activity (in the absence of photoreceptors) that also includes new microneuroma neuropil.

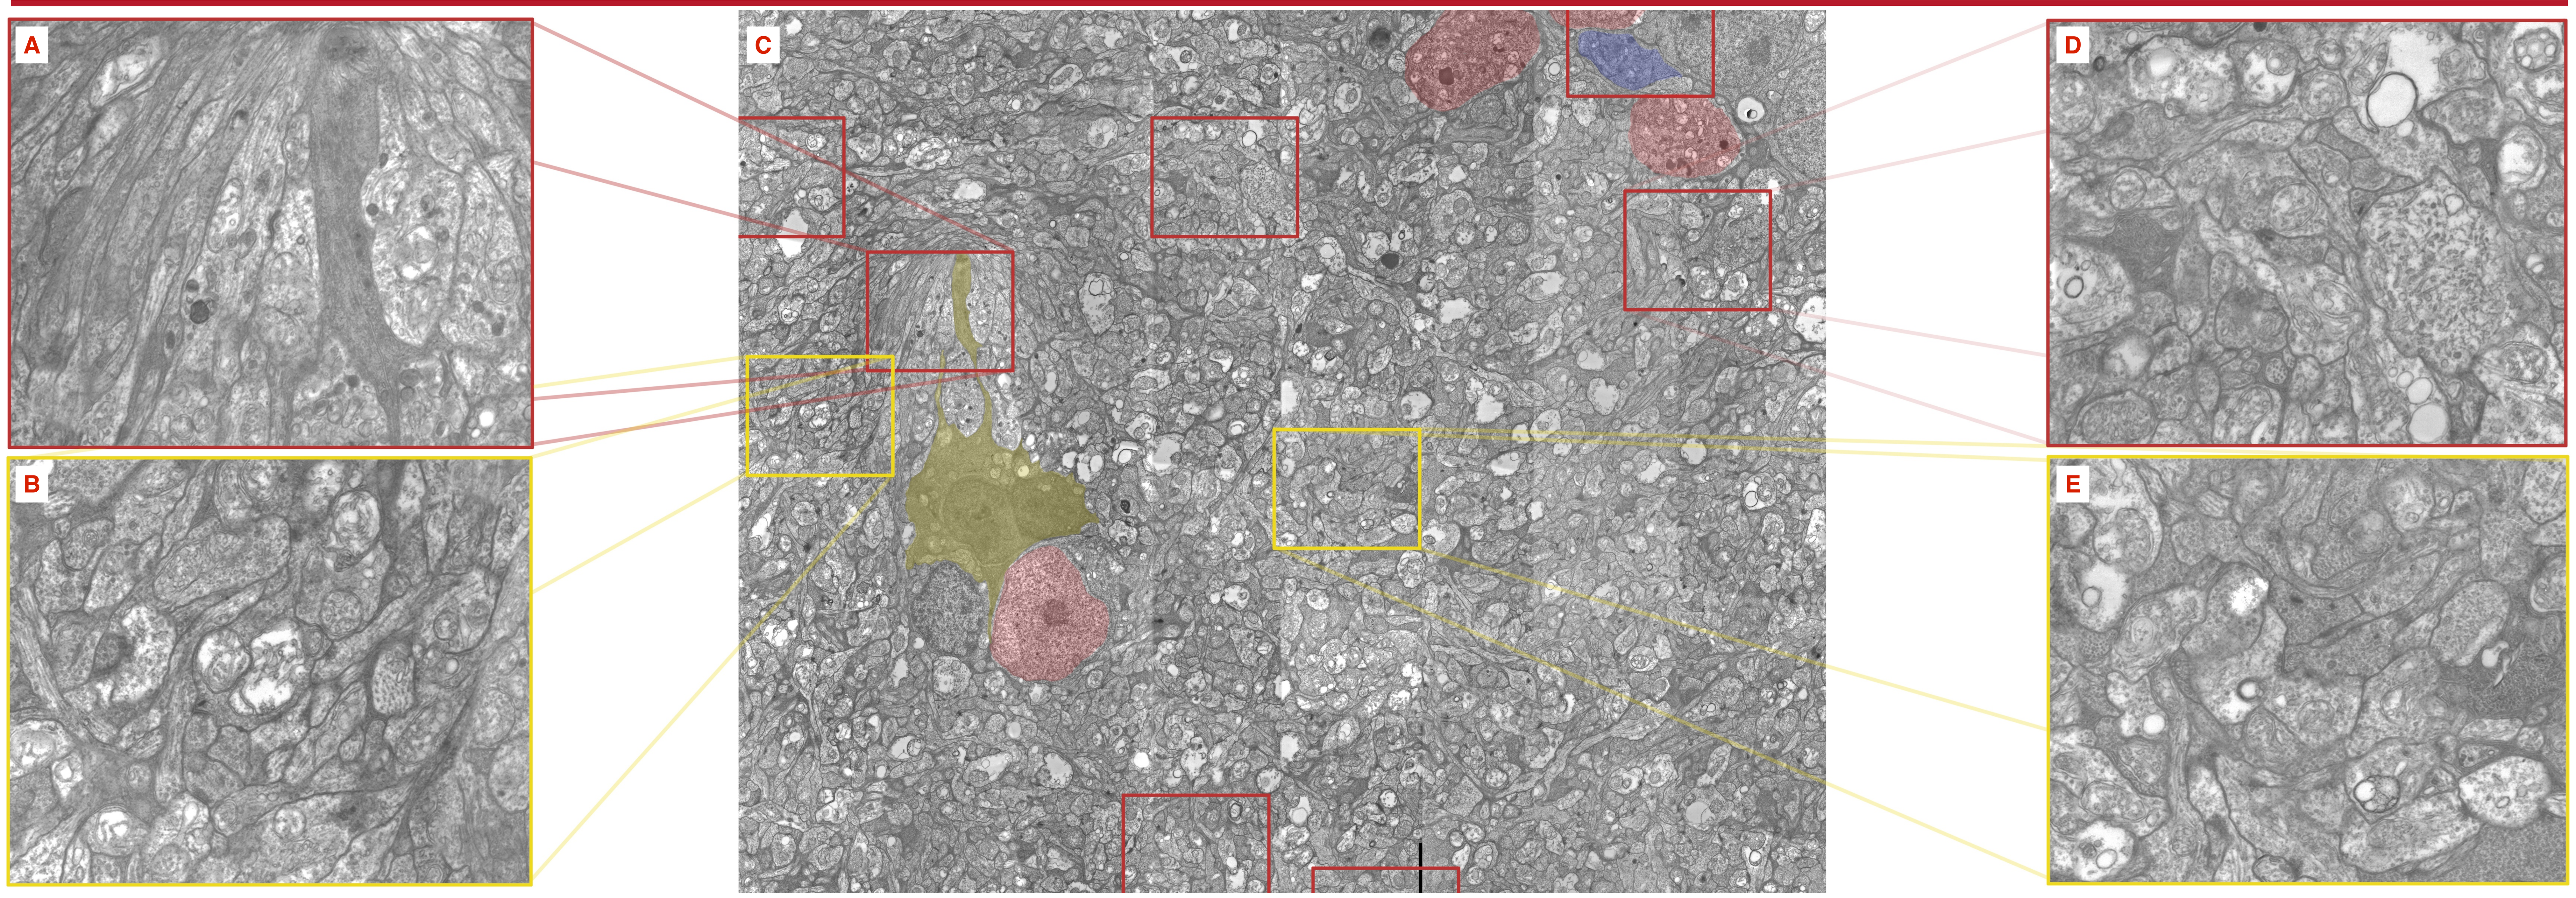
A
B
C
D
E

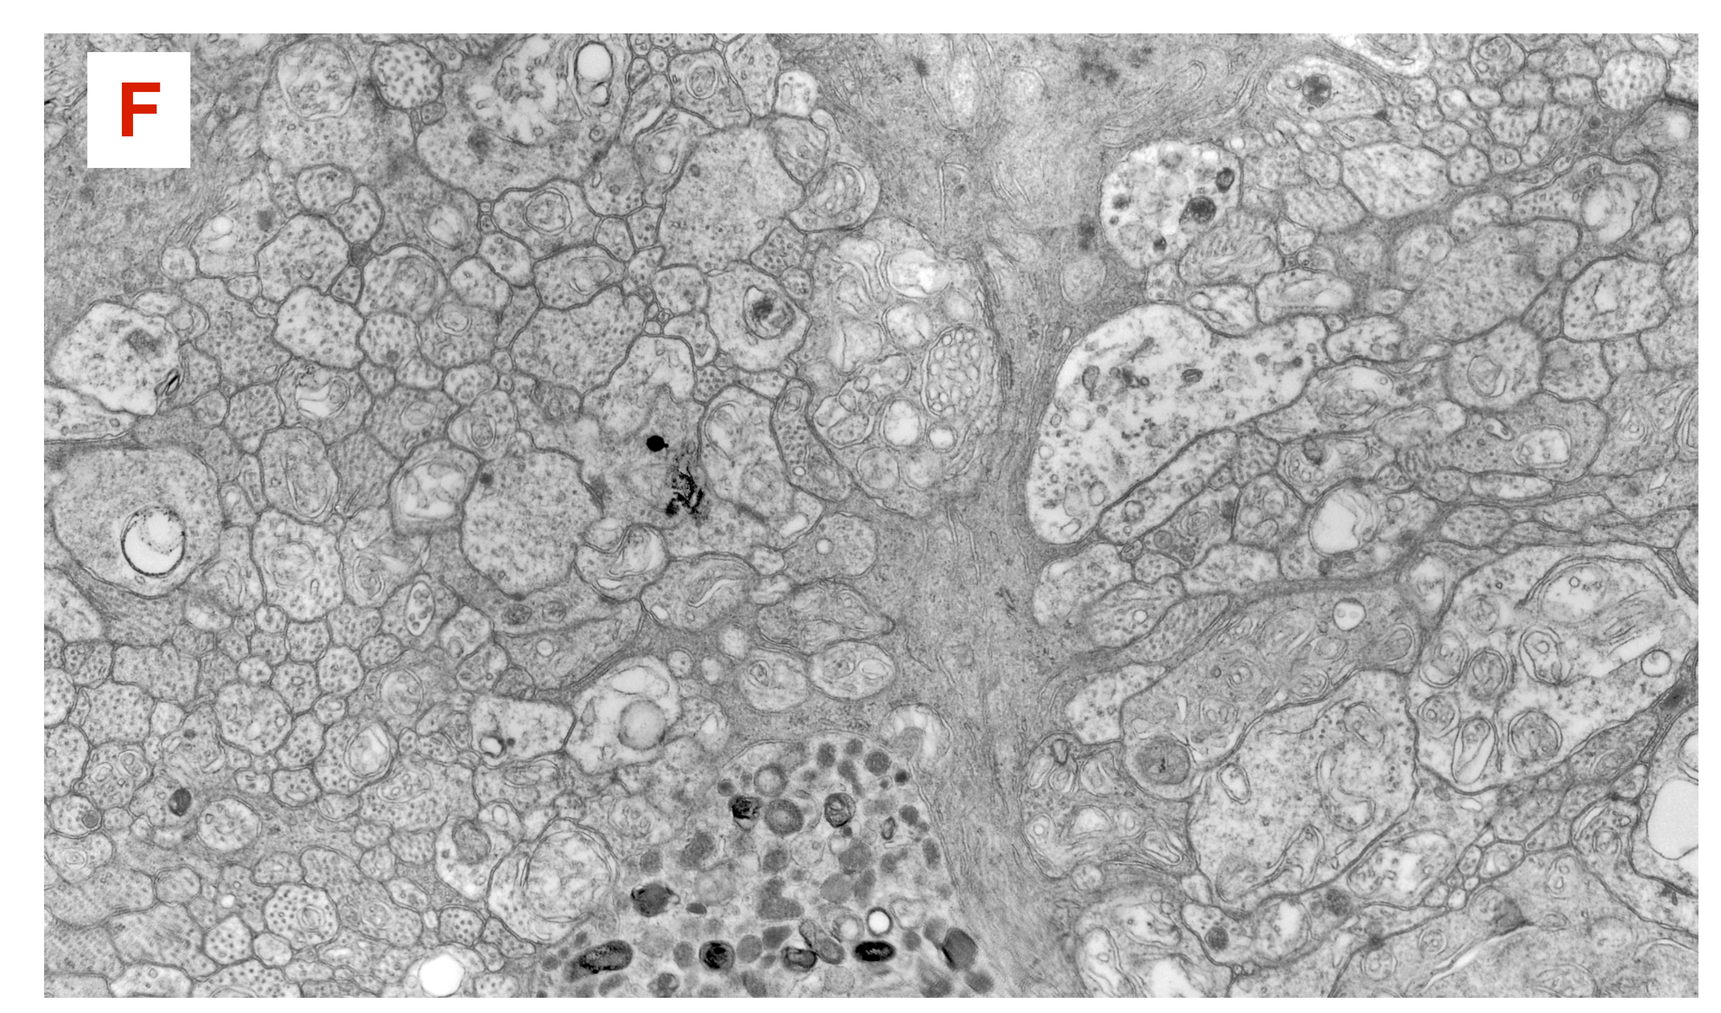
F

Figure 3. The above image composite [C] shows a section through a microneuroma in the TG9N RGS9 truncation mutant mouse (Jones et al. 2003), confirming the abundance of a variety of synapses implied by the AGB mapping experiments. Computational Molecular Phenotyping (CMP) fused with electron microscopy (Marc and Liu 2000) allows ultrastructural datasets to be combined with multiple probes of small molecules. This permits classification of participating cell classes found in microneuromas. The red cells in [C] are GABAergic amacrine cells. The blue tinted cell is a bipolar cell and the yellow tinted cell is a Müller cell. Both conventional [A] and ribbon synapses [B, D, E] are abundant and display all common synaptic arrangements (amacrine > amacrine, amacrine > bipolar, amacrine > ganglion cell, bipolar > amacrine, bipolar > ganglion cell) as well as instances of possible bipolar > bipolar contacts. This are likely fictive. Additionally, processes from retinal pigment epithelial (RPE) cells can be seen in association with some microneuromas, with the process in [F] projecting into the core of the microneuroma. While this reorganized neuropil contains sterotypical structural components, modeling of reconstructed circuits, as well as excitation mapping argue that these new wirings likely produce oscillatory networks.

Further reconstruction of microneuromas is underway with novel code incorporating algorithms to deal with the nonlinear distortions inherent in electron microscopy. This will allow the complete reconstruction of neural structures with automated image mosaicking and slice to slice registration of terabyte sized datasets.