Bipolar Cell “Accessory ON” Input to Amacrine Cells in the OFF Inner Plexiform Layer

Authors • J. Scott Lauritzen, Bryan W. Jones, Carl B. Watt, Shoeb Mohammed, James R. Anderson, Robert E. Marc
Moran Eye Center, University of Utah, Salt Lake City, UT

Grants • NIH EY02576 (RM), NIH EY015128 (RM), NSF 0941717 (RM), NIH EY014800 Vision Core (RM), RPB award to Moran Eye Center, RPB Career Development Award (BWJ), Thome Foundation Grant for AMD Research

Purpose: Recent confocal studies report axonal ribbon synapses from inner plexiform layer (IPL) 4/5-stratifying ON Cone Bipolar Cells (ON CBCs) onto tyrosine hydroxylase-positive cells (TH1s), intrinsically photosensitive ganglion cells (ipRGCs) and bi-stratified diving ganglion cells (bsdGCs) in the OFF IPL of mammalian retina, thus comprising an "accessory on" input to the OFF layer. Our goal was to identify additional bipolar cell (BC) types suspected to participate in this accessory ON input, confirm the presence of BC axonal synapses with transmission electron microscopy (TEM), and identify the targets of such synapses.

Methods: BC networks in the ultrastructural rabbit retinal connectome RC1 were annotated with the Viking viewer, and explored via 3D rendering and graph visualization of connectivity (Anderson et al. 2011 The Viking viewer for connectomics: scalable multi-user annotation and summarization of large volume data sets. JMicroscopy: doi:10.1111/j.1365-2818.2010.03402.x). Small molecule signals embedded in RC1, e.g. 4-aminobutyrate (γ), glycine (G), and L-glutamate (E), combined with morphological reconstruction allow for robust BC classification.

Results: Glycine positive bipolar cells (GBCs) with CBb3, CBb3-4, CBb4, CBb5, and Wide-Field morphology (MacNeil et al., 2004 classification scheme) form axonal ribbon synapses in IPL OFF layers, targeting glycinergic amacrine cells (GACs) and GABA-ergic amacrine cells (γ ACs). The targeted GACs synapse onto cells of mixed or opposite ON-OFF polarity. TEM ultrastructure confirms the presence of pre- and post-synaptic components necessary for functional synapses at the observed locations. Additional axonal ribbon synapses were confirmed with RGCs, as yet unidentified ACs, and unknown targets, which may be the TH1s, ipRGCs, and bsdGCs reported in the previous literature. Thus our results are consistent with and extend previous observations.

Conclusions: Multiple ON CBC types violate the IPL stratification rules, demonstrating a richer accessory ON network architecture than previously reported. γ AC and GAC targets of axonal synapses in the OFF layer represent novel targets of this accessory ON input. GAC targets mediate crossover inhibition via accessory ON pathways, revealing novel configurations for these functional networks.

Commercial Relationship: JS Lauritzen, None; BW Jones, None; CB Watt, None; S Mohammed, None; JR Anderson, None; RE Marc, Signature Immunologics

Figure 1. Multiple ON CBC Types Form Axonal Ribbons in the OFF IPL, Comprising an “Accessory ON” Input to the OFF Layer

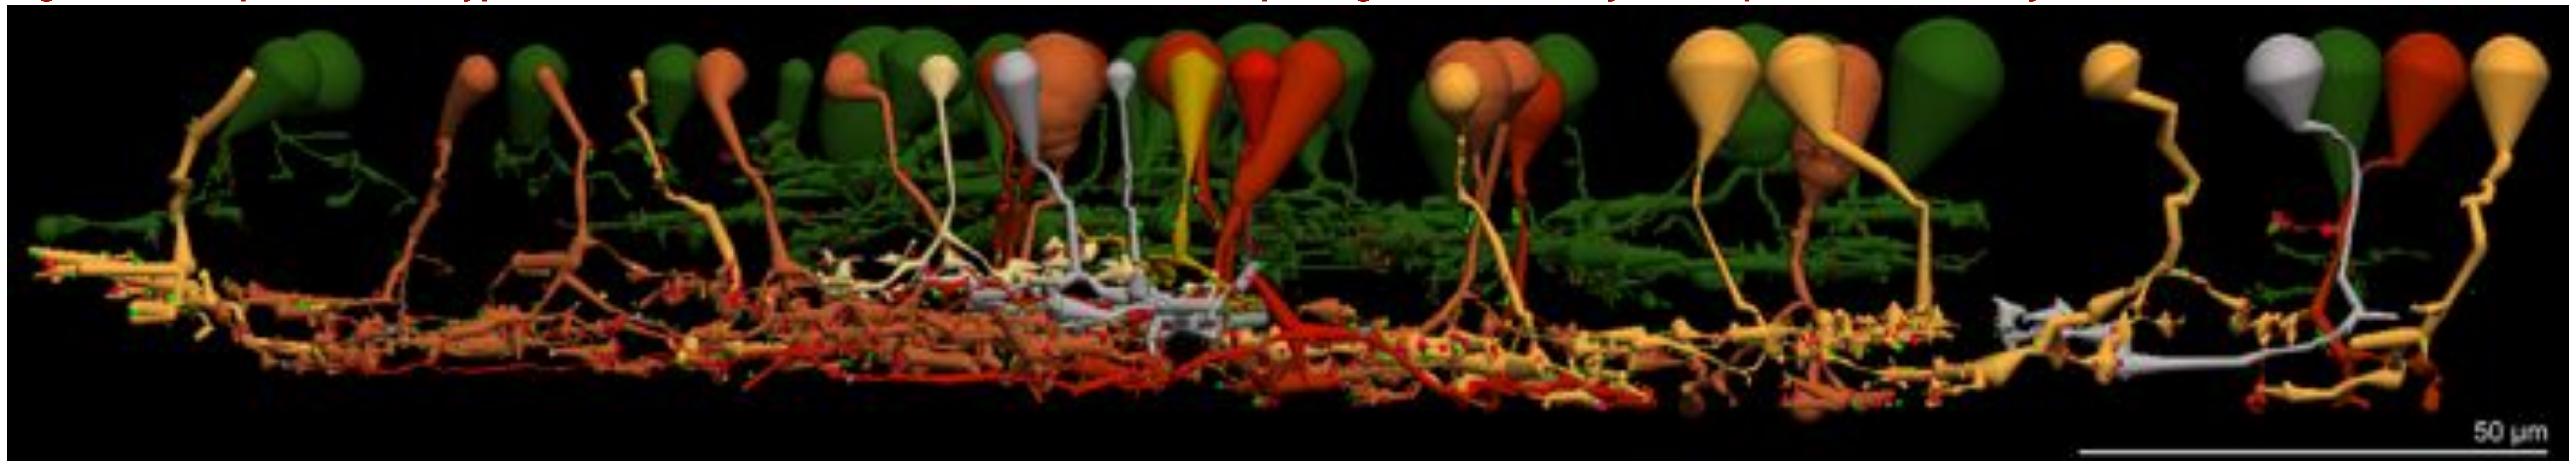
50 µm

Computational reconstruction of BCs reveals axonal ribbons formed by CBb3, CBb3-4, CBb4, CBb5, CBb6, & WFBCs (neutral/warm colors) in the OFF layers of the IPL. CBa1, CBa1-2, & CBa2 (green) demarcate the OFF IPL. Ribbon synapses shown as green dots. Post-synaptic densities shown as red dots.

Figure 2. Glycine Profiles & AII Coupling Confirm ON CBC Identities

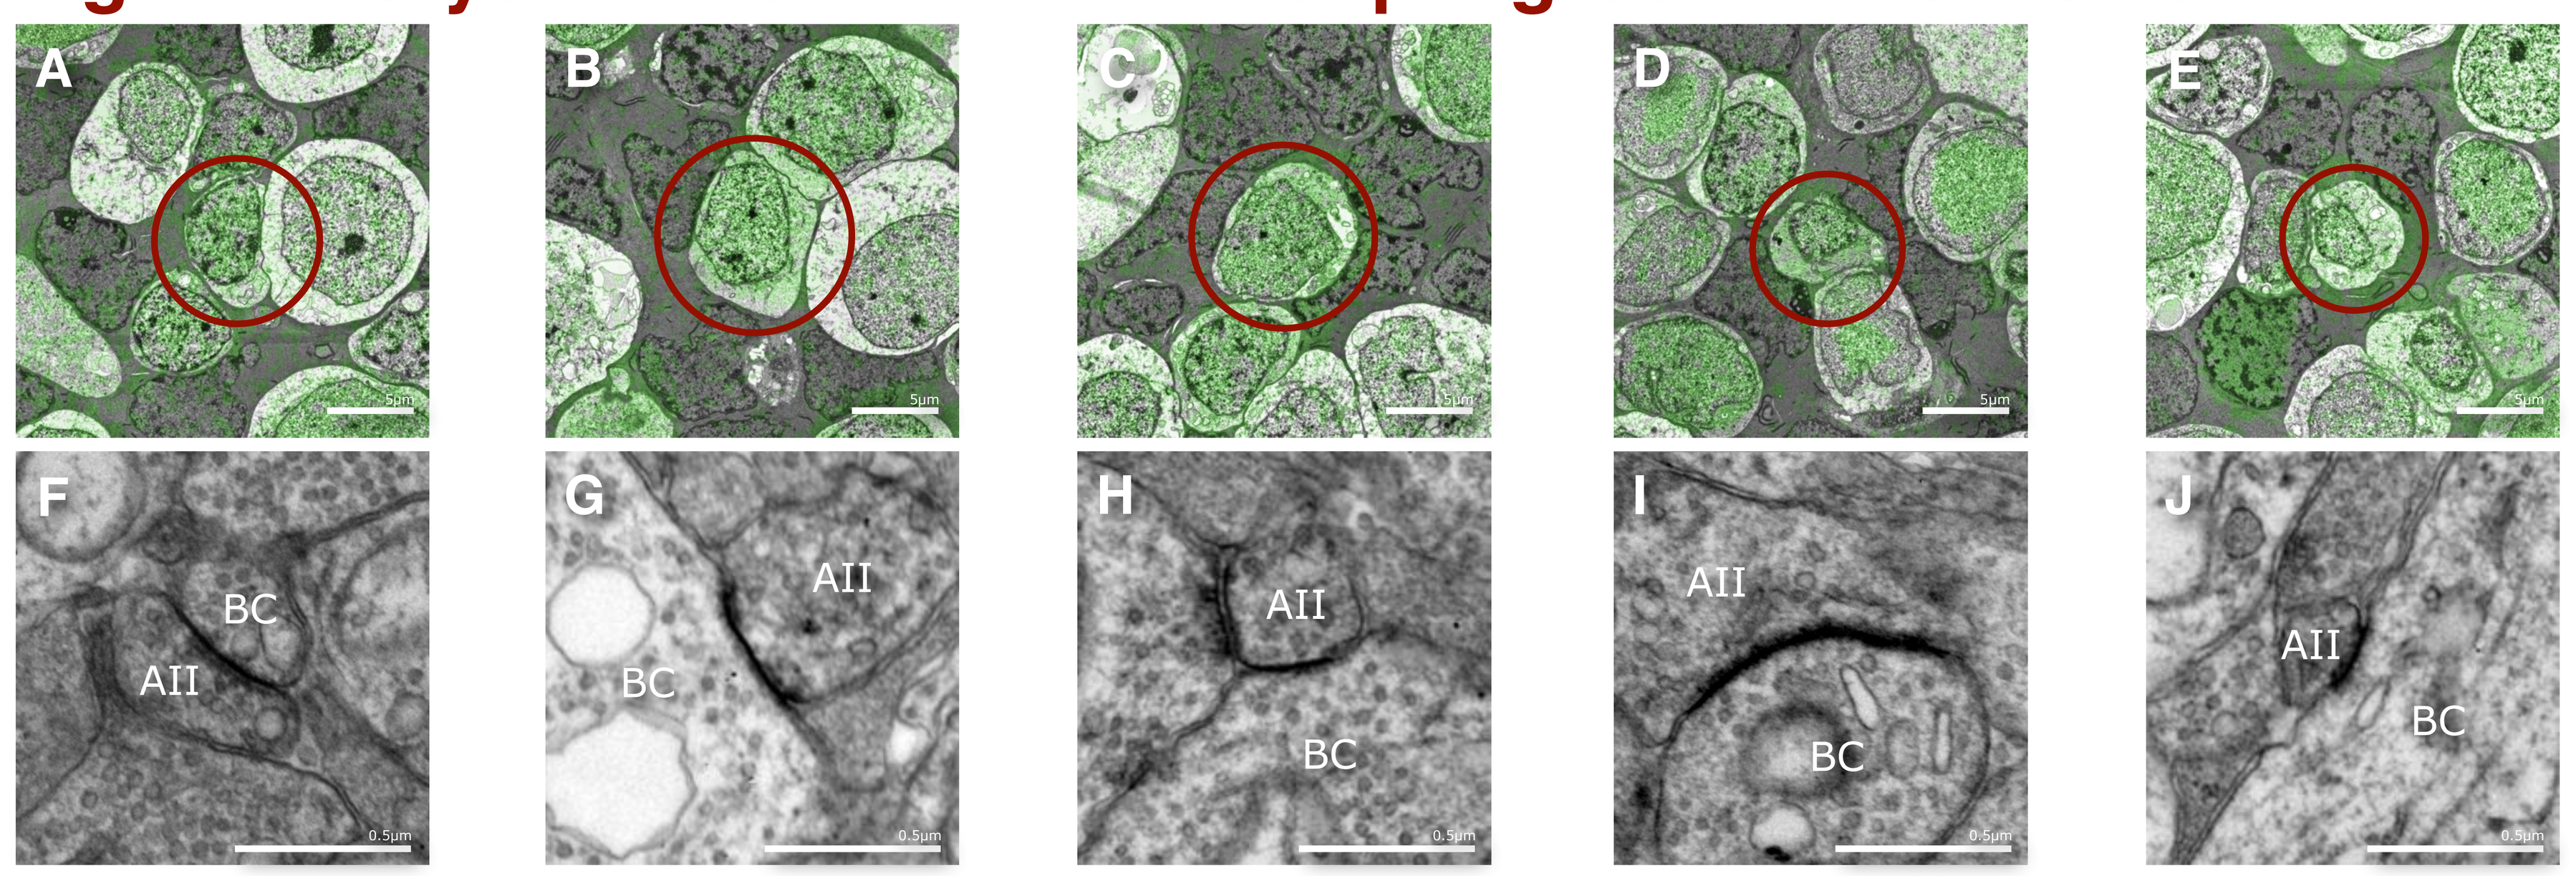
A
B
C
D
E
F
BC
AII
G
AII
BC
H
AII
BC
I
AII
BC
J
AII
BC

Examples of ON CBCs confirmed as glycine-positive (top row) & coupled to AII ACs (bottom row). **A,F.** CBb3-4. **B,G.** CBb5. **C,H.** CBb6. **D,I.** WFBC. **E,J.** Giant WFBC.

Figure 3. RGC Targets

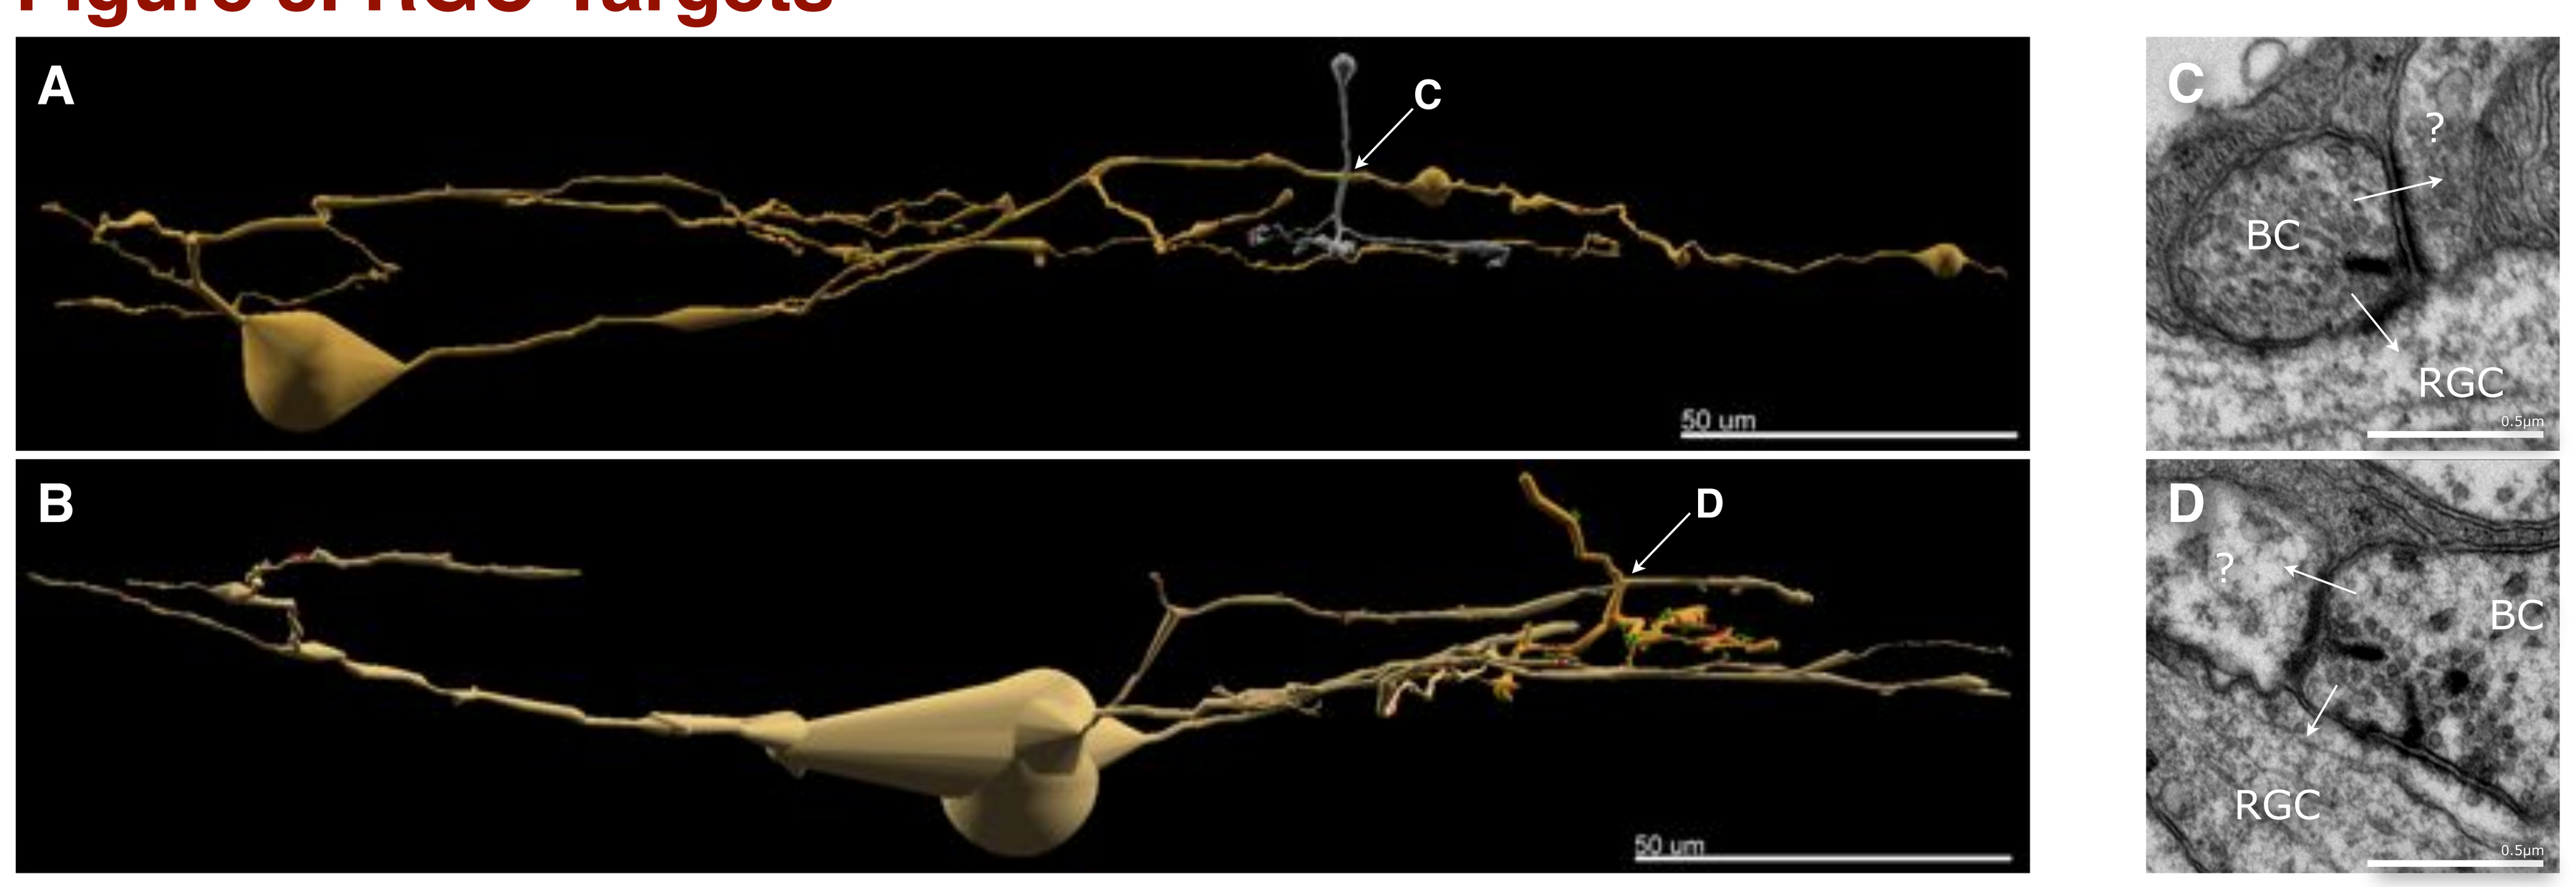
A
C
50 um
B
D
50 um
C
BC
?
RGC
D
?
BC
RGC

A,B. A CBb4 & CBb5, respectively, each form axonal ribbons onto a RGC. **C,D.** TEM confirmation of axonal ribbon diads formed by the BCs in A & B. White arrows indicate synapse directionality.

Figure 4. GAC Targets

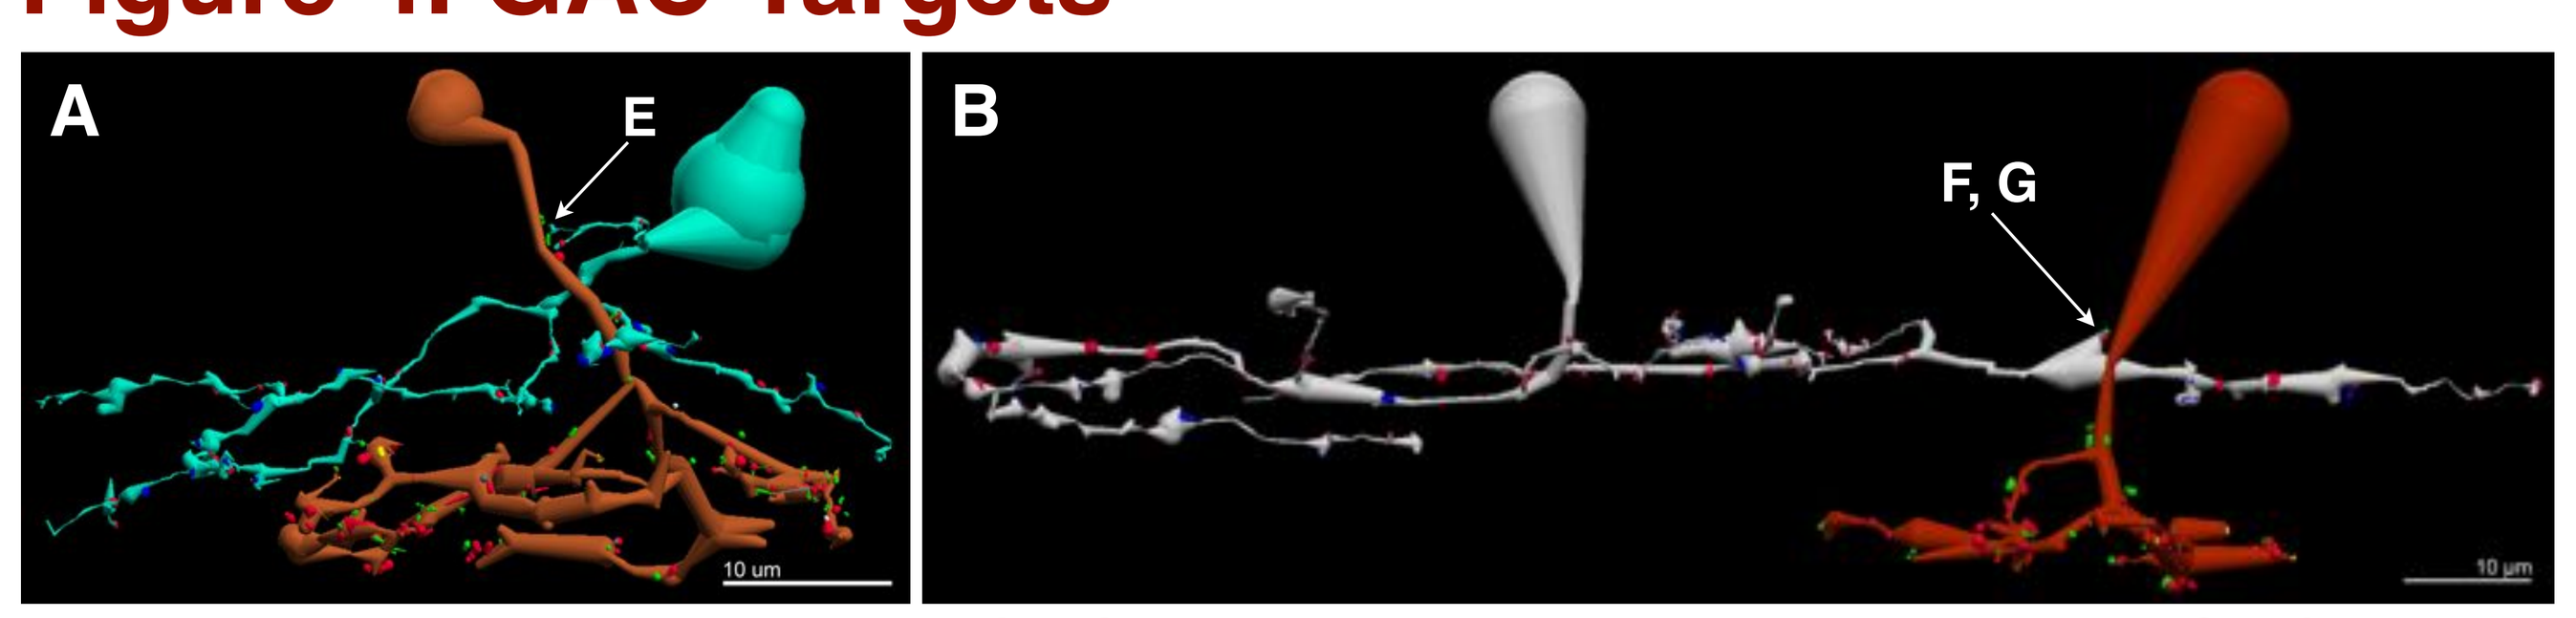
A
E
10 um
B
F, G
10 um

A,B. CBb5 & CBb6 axonal ribbons target multi- & mono-stratified GACs. **C,D.** Glycine-positive signatures of the GACs in A & B, respectively.

E,F. Axonal ribbons onto the GACs in panel A & B, respectively. **G.** Conventional synapse between the GAC & the descending axon of CBb6 in B. **E-G.** White arrows indicate synapse directionality.

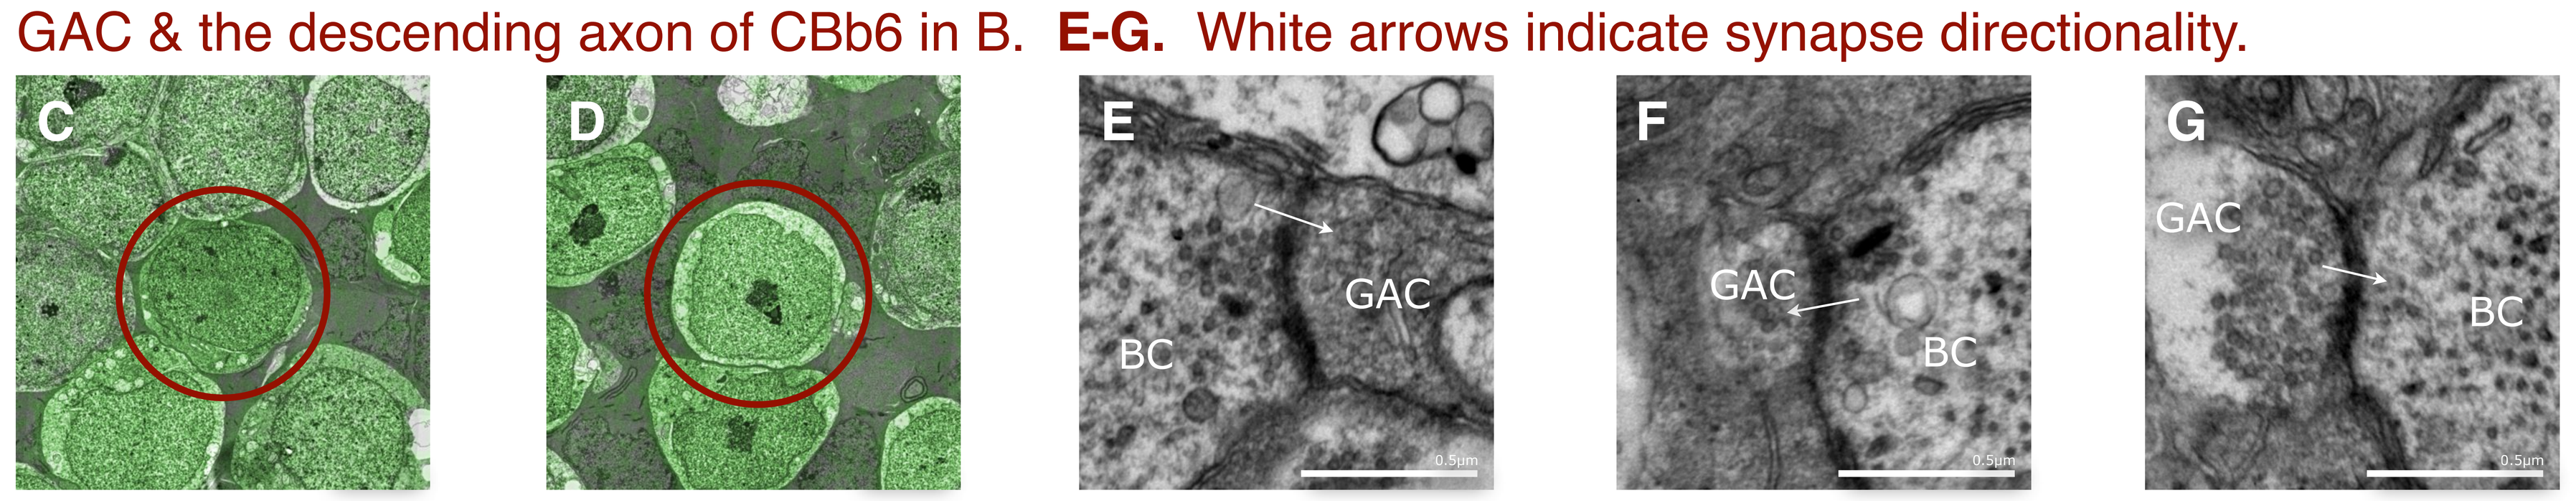
C
D
E
GAC
BC
F
GAC
BC
G
GAC
BC

Figure 5. γ AC & RGC Shared Targets, Other AC Targets, BC Axonal Cisterns

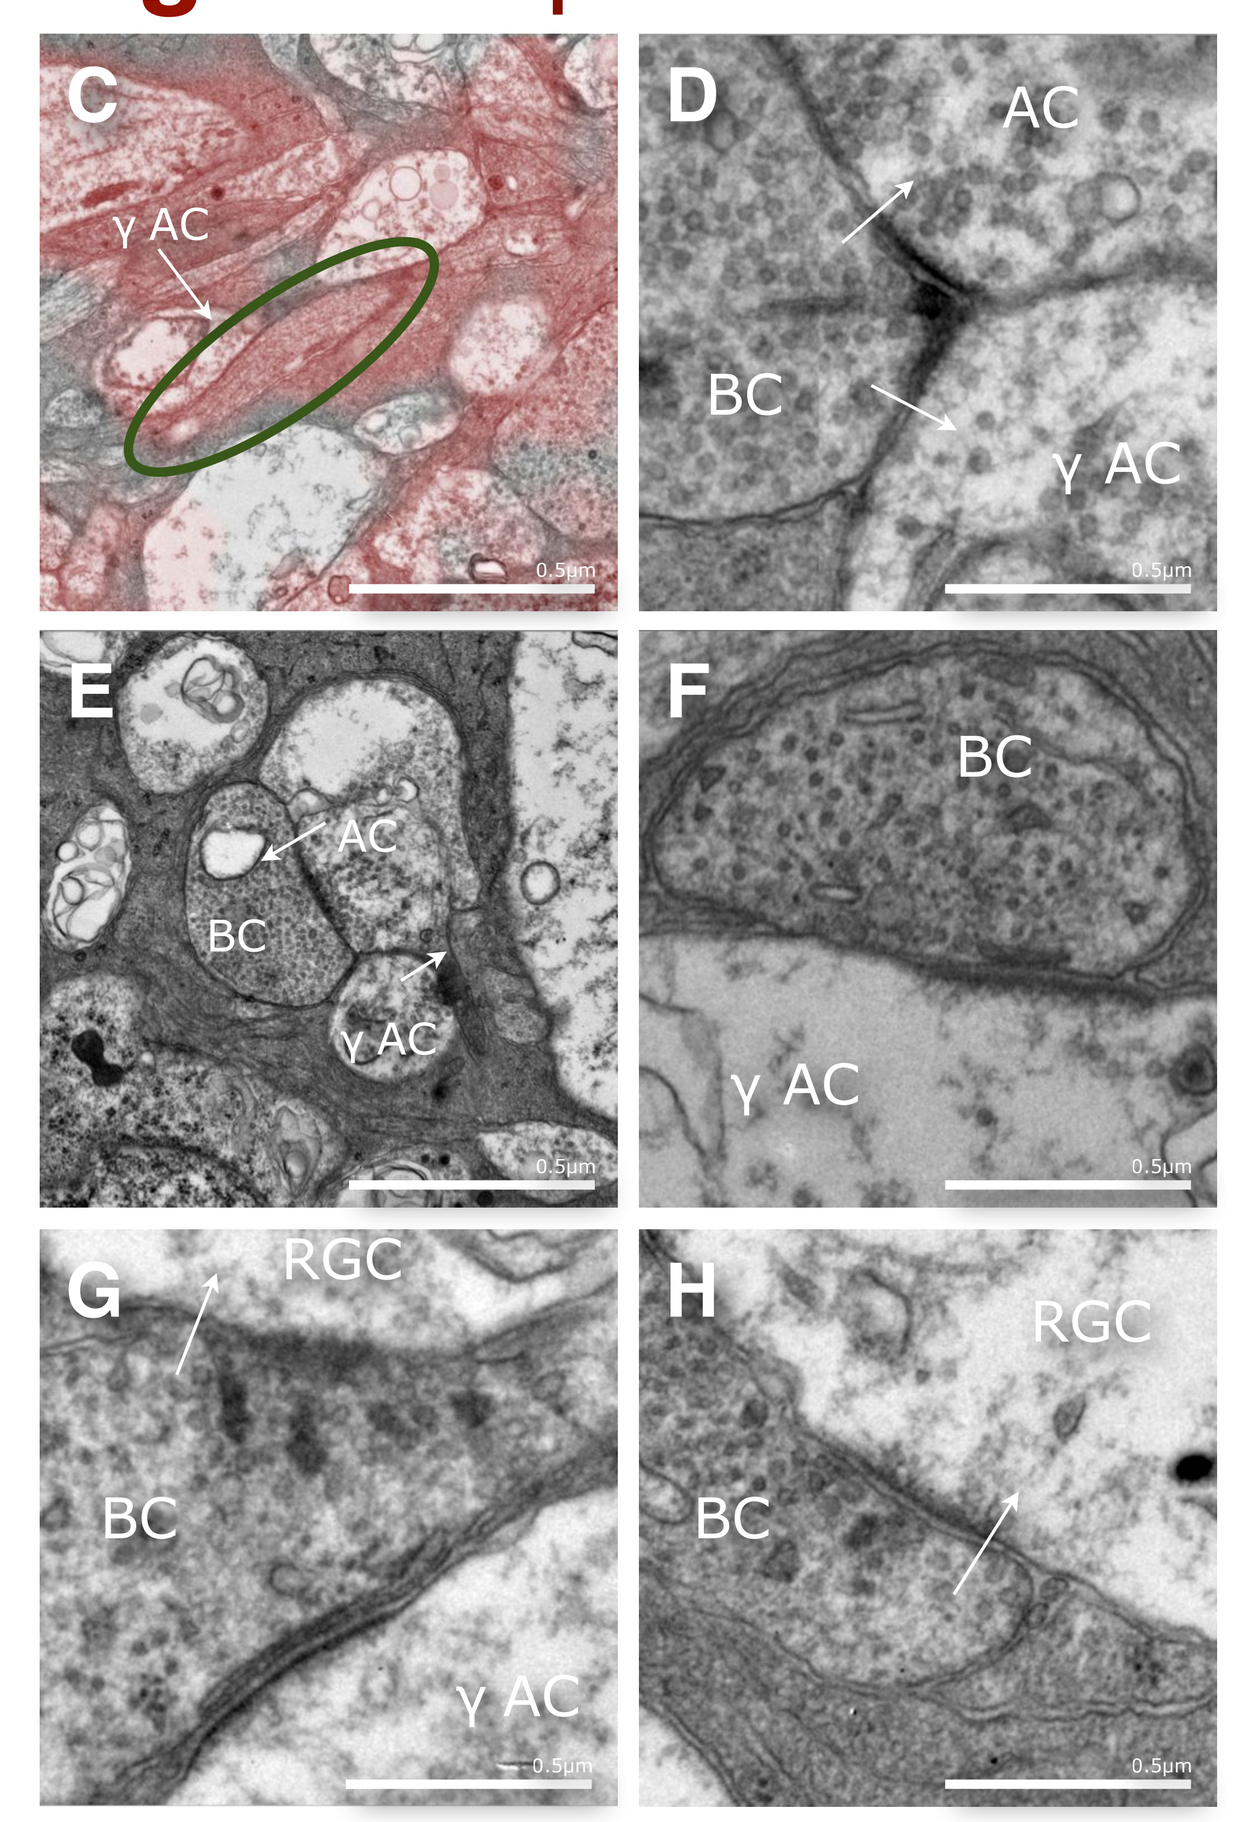
C
γ AC
D
AC
BC
γ AC
E
AC
BC
γ AC
F
BC
γ AC
G
RGC
BC
γ AC
H
RGC
BC

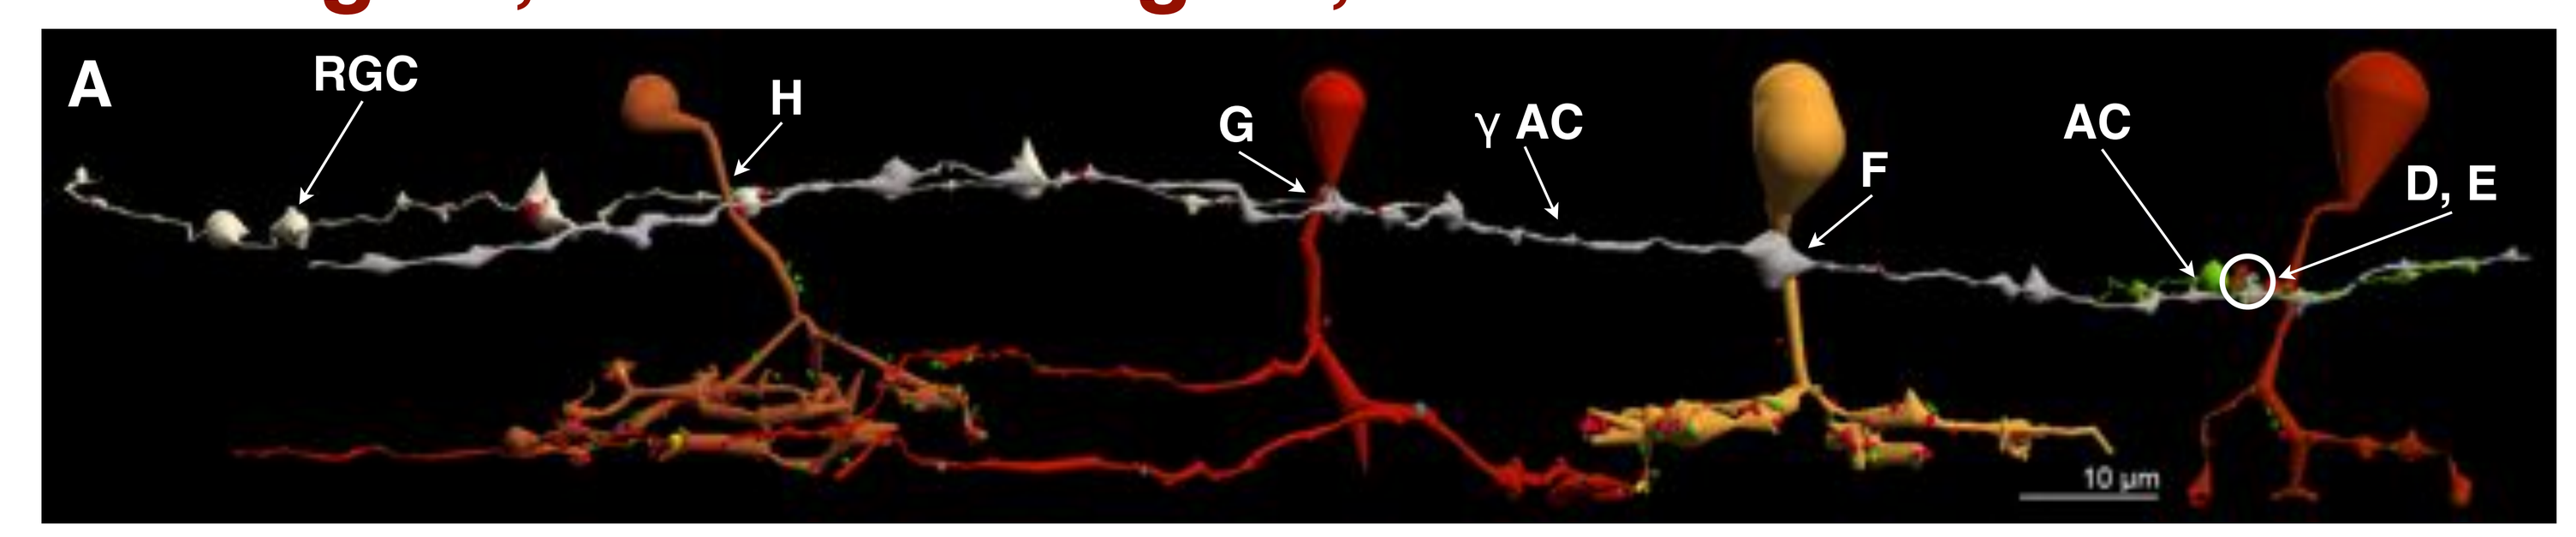
A
RGC
H
G
γ AC
F
AC
D, E
10 µm

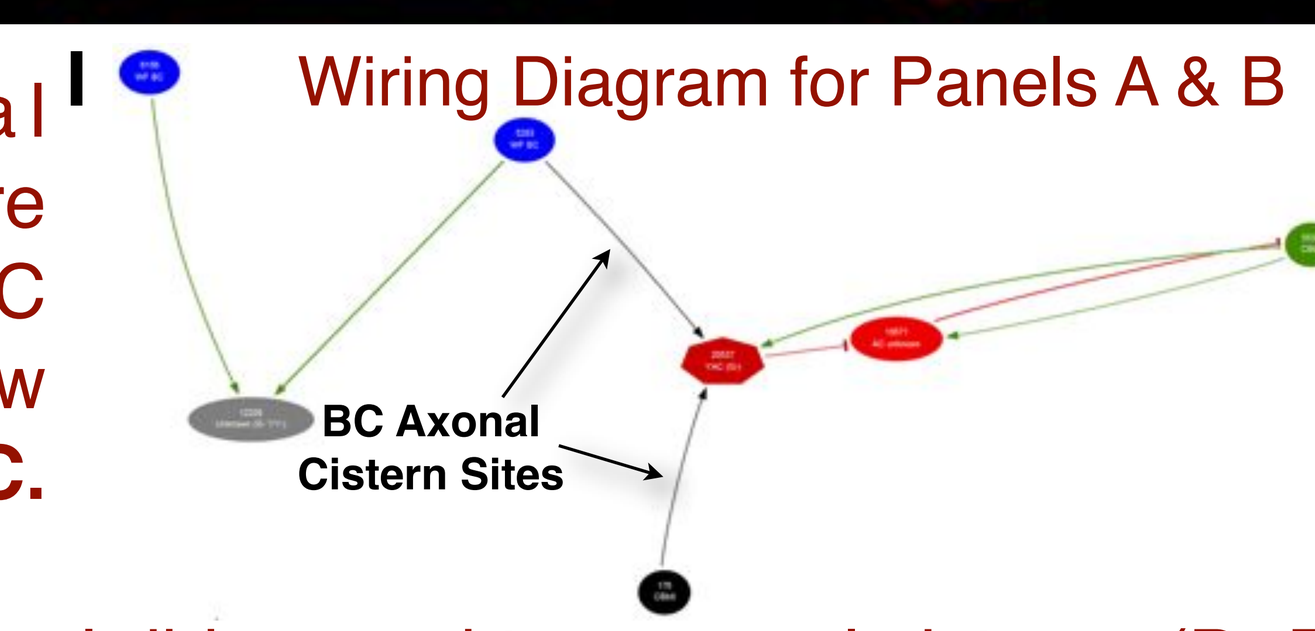
I
Wiring Diagram for Panels A & B
BC Axonal
Cistern Sites

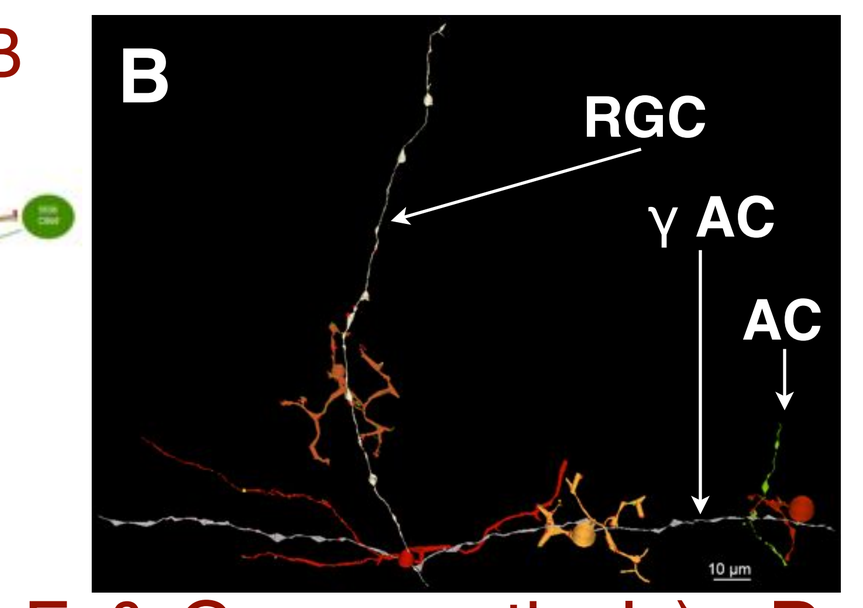
B
RGC
γ AC
AC
10 µm

A. ON CBC axonal ribbons & cisterns share common RGC & γ AC targets. **B.** Horizontal view of the network in A. **C.** GABA-positive signature of AC recipient to an axonal ribbon and two axonal cisterns (D, F, & G, respectively). **D.** CBb6 axonal ribbon diad onto the γ AC & other AC in A. **E.** The other AC target is likely a γ AC as it is glycine-negative (data not shown) & participates in nested FB with the γ AC. **F.** CBb5 axonal cistern onto the γ AC target. **G.** A WFBC multi-ribbon axonal synapse onto a RGC & an axonal cistern onto the γ AC targeted in A, D, & F. **H.** Another WFBC forms an axonal ribbon onto the same RGC shown in G. **D-H.** White arrows indicate synapse directionality. **I.** Graph visualization of A-B.

Figure 6. Crossover Inhibition via Accessory ON Pathways

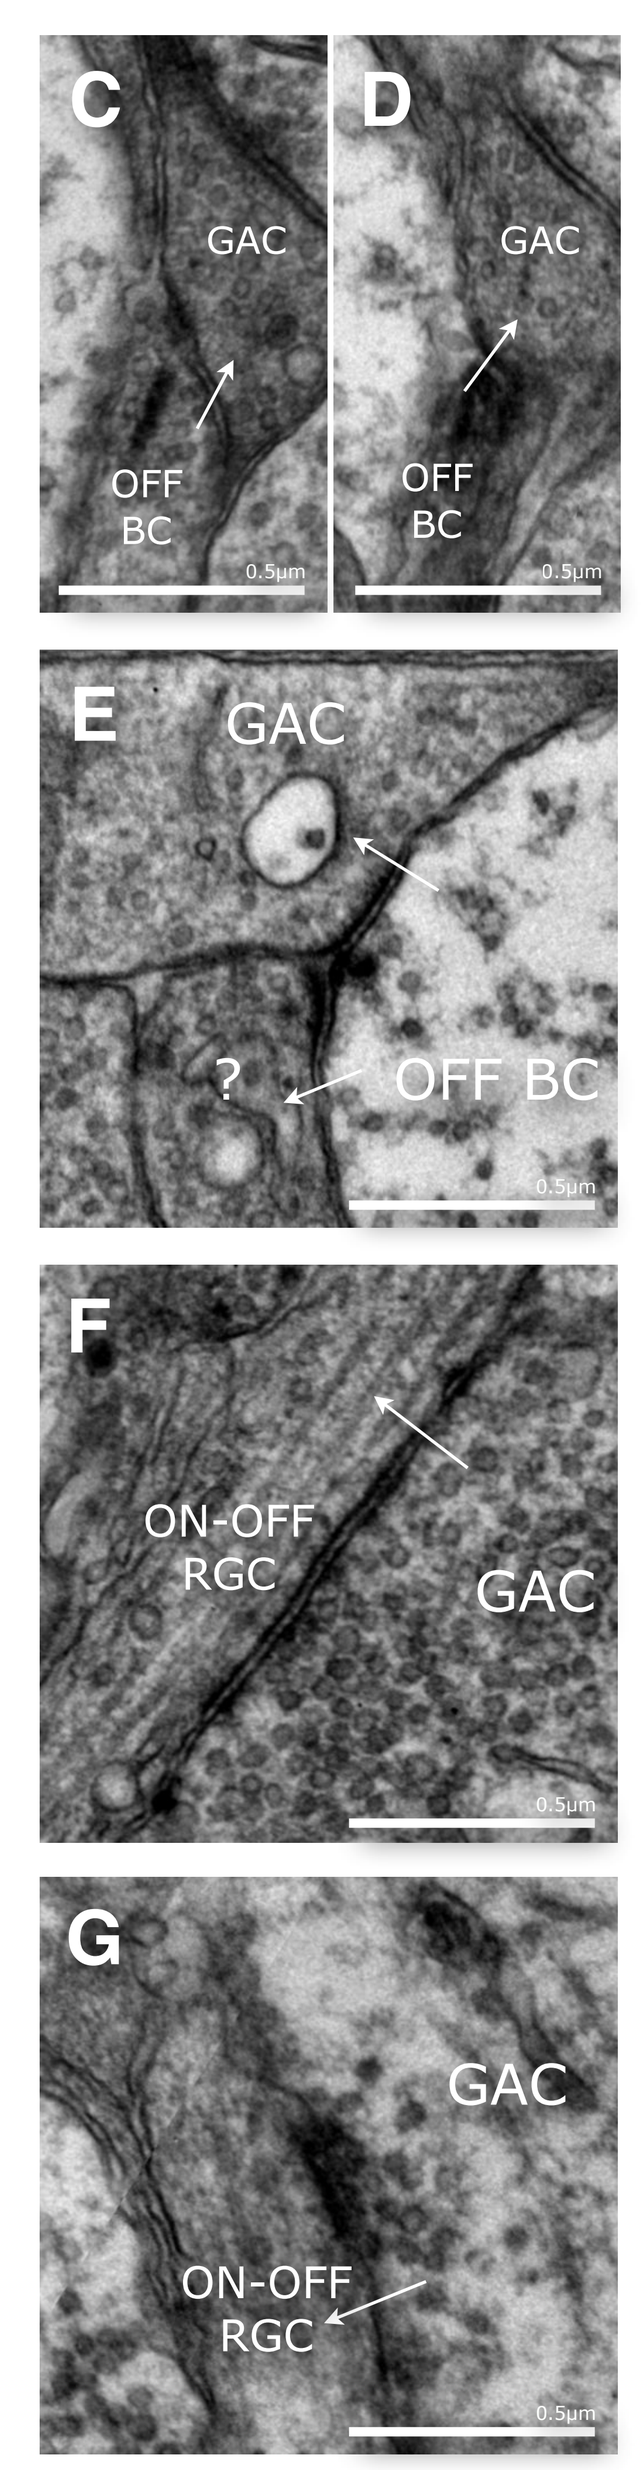
C
GAC
OFF BC
D
GAC
OFF BC
E
GAC
?
OFF BC
F
ON-OFF RGC
GAC
G
GAC
ON-OFF RGC

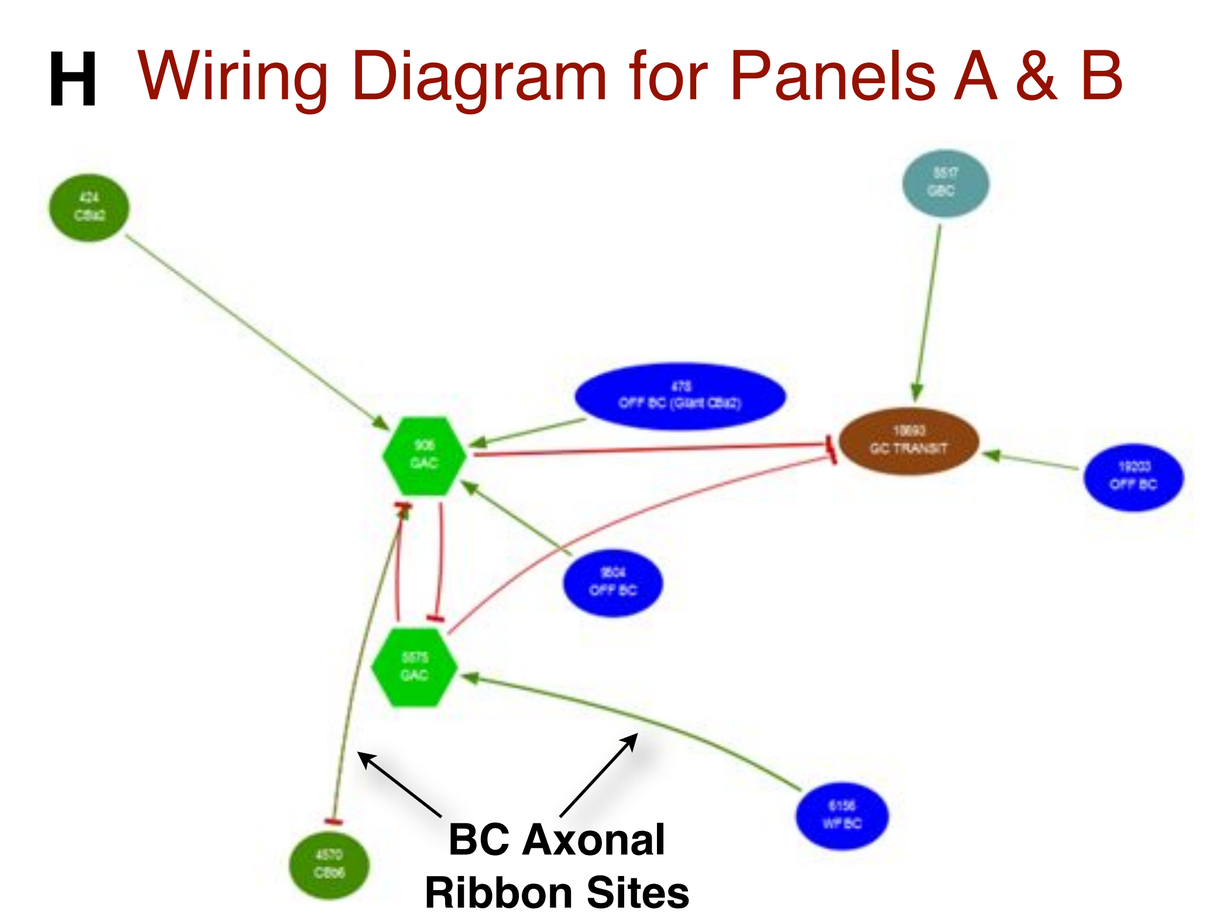
H Wiring Diagram for Panels A & B
BC Axonal
Ribbon Sites

BC-GAC-BC OFF-ON CROSSOVER INHIBITION

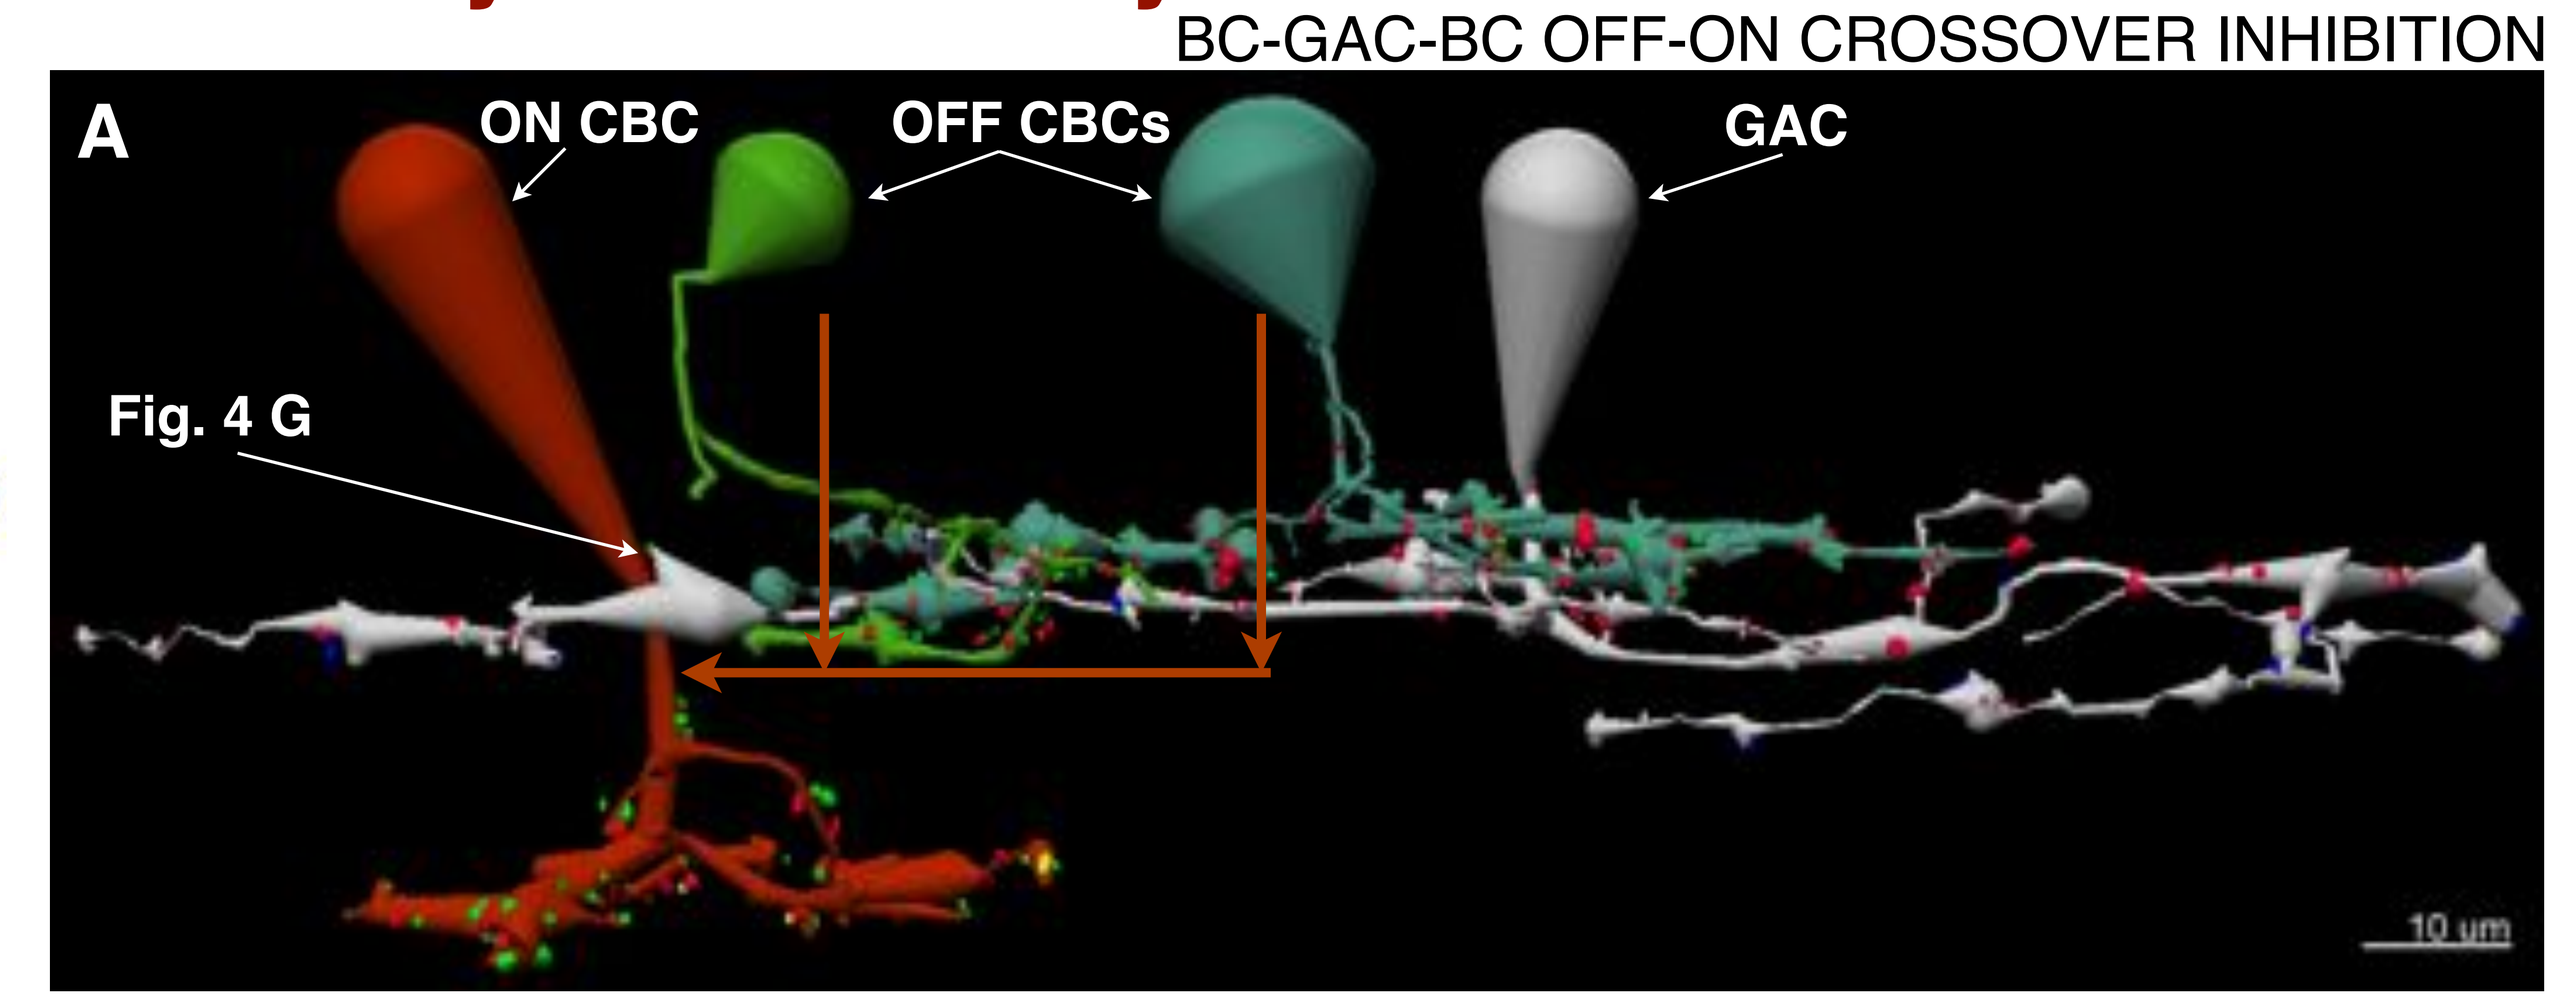
A
ON CBC
OFF CBCs
GAC
Fig. 4 G
10 um

A. Two CBa2s synapse onto a mono-stratified GAC, which synapses onto an ON CBC at a reciprocal synapse with an axonal ribbon, constituting accessory OFF input at accessory ON locations. **A-B.** Orange arrows represent the flow of information. **B.** Axonal ribbons from a CBb5 & CBb6 onto two GACs, each of which synapse onto a monostratified ON-OFF RGC. **C,D,E.** OFF BC ribbon synapses onto the GAC shown in panel A. **F,G.** GAC conventional synapses onto the RGC shown in panel B. **C-G.** White arrows indicate synapse directionality. **H.** Graph visualization of A-B.

BC-GAC-RGC ON-OFF CROSSOVER INHIBITION

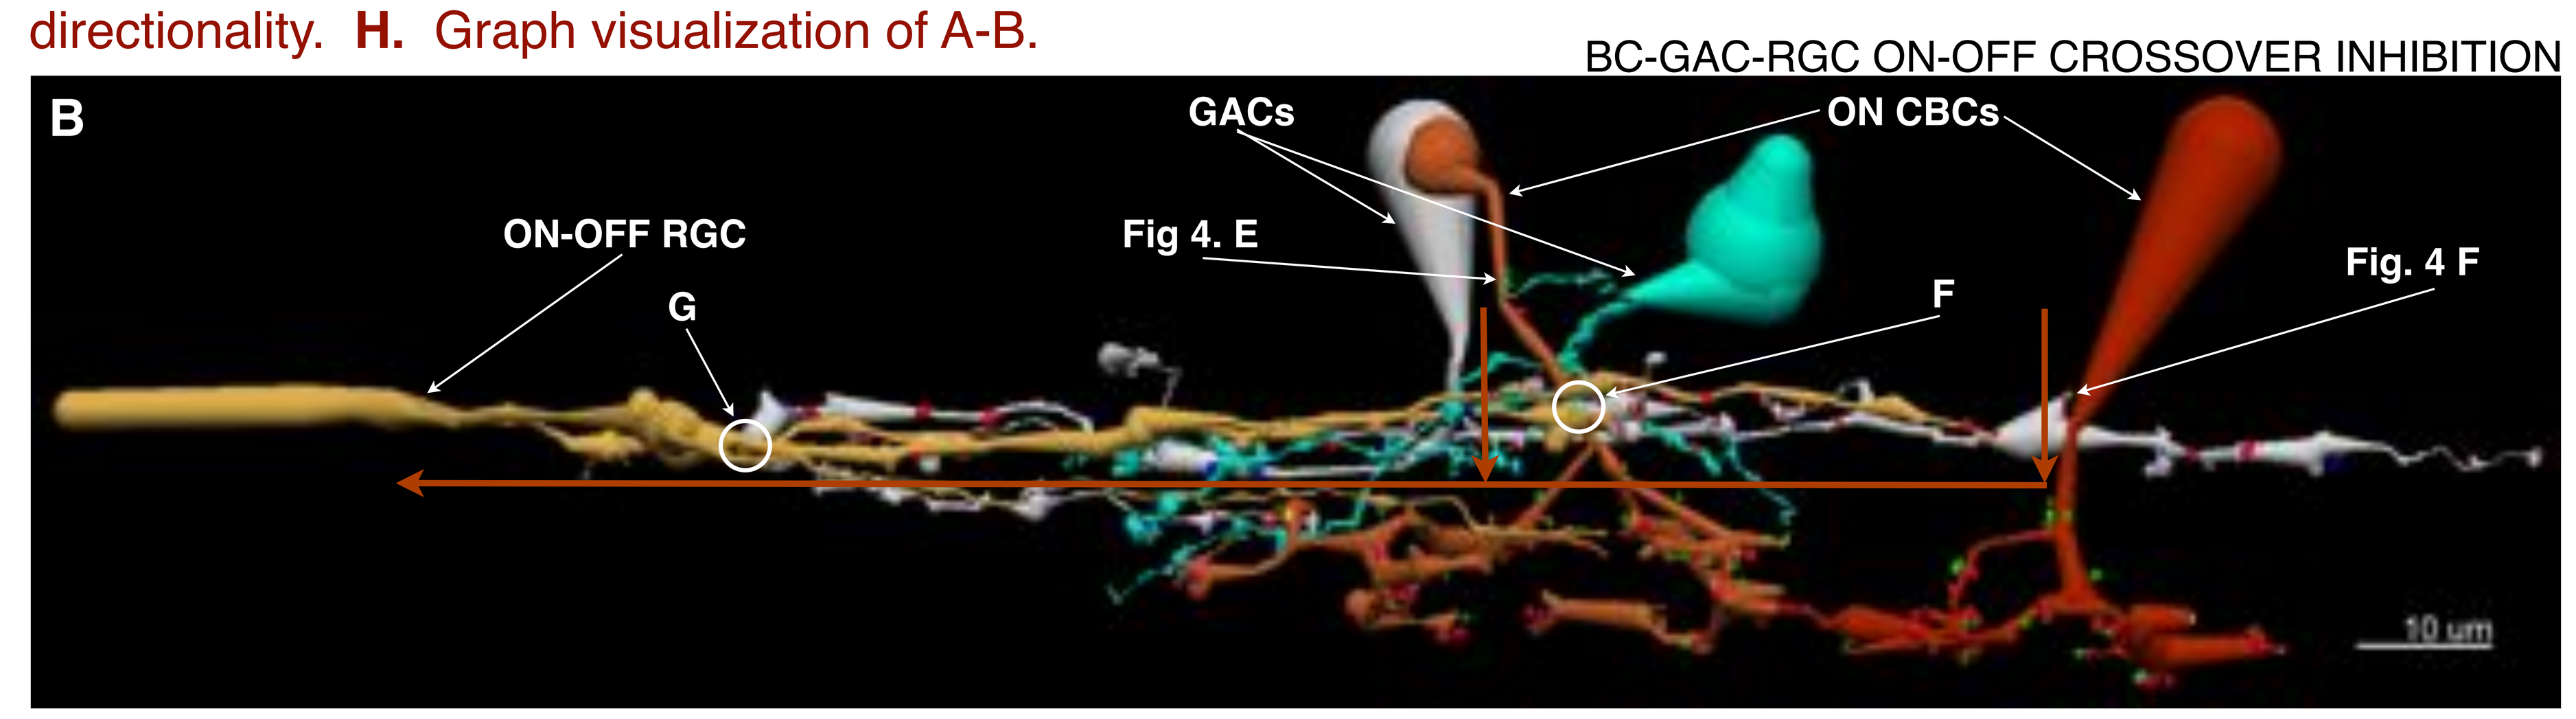
B
GACs
ON CBCs
ON-OFF RGC
G
Fig 4. E
F
Fig. 4 F
10 um